Tumor Microenvironment Pharmacodynamic Effect of Nemvaleukin Less Frequent Intravenous Dosing in Multiple Solid Tumors: Results From the Phase 1/2 ARTISTRY-3 Study

Sarah S. Donatelli,[1] Justin A. Call,[2] Alexander I. Spira,[3] Jorge Bartolomé,[4] Maria de Miguel,[5] Xiwei Chen,[1] Nehal J. Lakhani,[6] Sonali Panchabhai,[1] Sarina A. Piha-Paul[7]

[1]Mural Oncology, Waltham, MA, USA; [2]START Mountain Region, West Valley City, UT, USA; [3]NEXT Oncology Virginia, Fairfax, VA, USA; [4]UTEC Hospital Clínico San Carlos, Madrid, Spain; [5]START Madrid-CIOCC, Centro Integral Oncológico Clara Campal, Madrid, Spain; [6]START Midwest, Grand Rapids, MI, USA; [7]Department of Investigational Cancer Therapeutics, Division of Cancer Medicine, The University of Texas MD Anderson Cancer Center, Houston, TX, USA

Take Away

- Less frequent IV dosing of nemvaleukin demonstrated tumor-site immune activation
- A shift toward a proinflammatory immune milieu dominated by cytolytic effector NK cells and CD8$^+$ T cells was observed upon nemvaleukin treatment
- These findings are consistent with the peripheral immune dynamics and proposed mechanism of action of nemvaleukin[2,6]

Conclusions

- Deep immune profiling of paired tumor biopsies demonstrated immune activation with less frequent dosing of nemvaleukin
- Increased on-treatment density of cytolytic effector lymphocytes (GZB$^+$ NK and GZB$^+$ CD8$^+$ T cell subsets) was observed
- Ratios of inflammatory/suppressive immune cells (NK:T$_{reg}$ and CD8$^+$:T$_{reg}$) were favorable for the nemvaleukin less frequent dosing regimen
- Collectively, LFIV nemvaleukin resulted in site-specific pharmacodynamic activity and immune activation, with a shift toward proinflammatory milieu in heavily pretreated patients with advanced solid tumors
- These results were seen in biopsies from advanced solid tumors, including both MM and OC tumors, and are hypothesis-generating that nemvaleukin may have the potential to recruit cytolytic effectors to poorly immunogenic tumor sites
- There are limitations to the study. For example, the study population was heterogenous for tumor types and heavily pretreated for advanced disease. This study also compared effects of nemvaleukin in TME between patients from different schedules and tumors biopsied on different days after nemvaleukin treatment

Disclosures

SSD, XC: employment and stock options: Mural Oncology. **JAC, JB:** nothing to disclose. **AIS:** leadership role: NEXT Oncology Virginia (institution); honoraria, CytomX Therapeutics, AstraZeneca/MedImmune, Merck, Takeda, Amgen, Janssen Oncology, Novartis, BMS, Bayer, Prelude Therapeutics, AbbVie, Astellas Pharma; consulting or advisory boards: Incyte, Amgen, Novartis, Mirati Therapeutics, Gritstone Oncology, Jazz Pharmaceuticals, Takeda, Janssen, Mersana, Gritstone Bio, Daiichi Sankyo/AstraZeneca, Regeneron, Lilly, Black Diamond Therapeutics, Sanofi, ArriVent Biopharma, Synthekine, GSK, CRISPR Therapeutics; funding to the institution: AstraZeneca/MedImmune, Merck, BMS, Blueprint Medicines, Roche, AstraZeneca, Boehringer Ingelheim, Astellas Pharma, MedImmune, Novartis, Incyte, Abbvie, Ignyta, Takeda, Macrogenics, CytomX Therapeutics, Astex Pharmaceuticals, BMS, Loxo, Gritstone, Plexxikon, Amgen, Daiichi Sankyo, ADCT, Janssen Oncology, Rubius, Synthekine, Mersana, Blueprint Medicines, Regeneron, Alkermes, Revolution Medicines, Medikine, Black Diamond Therapeutics, BluPrint Oncology, Nalo Therapeutics, Scorpion Therapeutics, ArriVent Biopharma, Revolution Medicines, Prelude Therapeutics; research funding: LAM Therapeutics. **MdM:** grants: AbbVie, Array, Achilles, Bayer, Basilea, Biontech, Faron, MSD, Novartis, Cytomex, Genentech, Genmab, Janssen, Menarini, Nektar, Catalym, Pharmamar, Regeneron, Zenith, Repair; consulting/advisory boards: Novartis, Syneoshealth, Repair. **NJL:** nonfinancial and other support: Alkermes/Mural Oncology during conduct of study; other support: ALX Therapeutics, Ascentage, Asana, BeiGene, Constellation Pharma/Morphosys, Alexion, Cerulean, Forty Seven, Alpine, Merck, Pfizer, InhibRx, Regeneron, Apexian, Formation Biologics (forbius), Symphogen, CytomX, Sapience Therapeutics, Incyte, Jounce, Livzon, Northern Biologics, Innovent Biologics; personal fees: Innovent Biologics, Ikena, Odonate , Loxo/Lilly, Alpine Biosciences, Ikena, Mersana, Macrogenics, Alpine, Helsinn, Seagen/Pfizer, Celgene, Samumed, Astellas, Shattuck Labs, SK Life Sciences, KSQ Therapeutics/Roche, Tizona, Servier, Sapience, Repare Therapeutics, Gilead, Bristol Myers/Celgene, GSK, Janssen, Artios, Arcus Bio, Volastra, Artios, Incyte, Revolution Medicines, Quanta Therapeutics, outside the presented work. **SP:** previous employment with and past stock options: Mural Oncology. **SAP-P:** research funding (institution): AbbVie, ABM Therapeutics, Acepodia Inc., Alkermes, Aminex Therapeutics, BioMarin Pharmaceutical, Boehringer Ingelheim, BMS, Cerulean Pharma Inc., Chugai Pharmaceutical Co., Cullinan Oncology Inc., Curis, Cyclacel Pharmaceuticals, Daiichi Sankyo, Eli Lilly, ENB Therapeutics, Epigenetix, Five Prime Therapeutics, F-Star Beta Ltd, F-Star Therapeutics, Gene Quantum, Genmab A/S, Gilead Sciences, GlaxoSmithKline, Helix BioPharma, Hengrui Pharmaceuticals, HiberCell, Immunomedics, Innovent Biologics Co., Incyte, Jacobio Pharmaceuticals, Jazz Pharmaceuticals, Jiangsu Simcere Pharmaceutical Co., Johnson & Johnson, Loxo Oncology, Lytix Biopharma, Medimmune, Medivation, Merck Sharp, and Dohme, Nectin Therapeutics, Novartis Pharmaceuticals, NRG Oncology, Nurix, OncoNano Medicine Inc., Pieris Pharmaceuticals, Pfizer, Phanes Therapeutics, Principia Biopharma, ProFoundBio US, Puma Biotechnology, Purinomia Biotech, Rapt Therapeutics, Replimune, Roche/Blueprint, Seattle Genetics, Silverback Therapeutics, Shasqi, Strand Therapeutics Inc., Synlogic Therapeutics, Taiho Oncology, Tallac Therapeutics Inc., Tesaro, Theradex Oncology, Toragen Therapeutics, TransThera Bio, Xencor, ZielBio, NCI/NIH, P30CA016672 – Core Grant (CCSG Shared Resources); consulting fees: CRC Oncology, Lilly USA, LLC.

Background

- Nemvaleukin alfa (nemvaleukin, ALKS 4230) is a novel, engineered cytokine designed to selectively bind to the intermediate-affinity interleukin-2 (IL-2) receptor and leverage the therapeutic benefits of the IL-2 pathway while mitigating the toxicities that historically restricted its clinical application[1]
- In the ARTISTRY-1 study, nemvaleukin showed antitumor activity as monotherapy and in combination with pembrolizumab in multiple solid tumors when administered intravenously (IV) at 6 µg/kg once daily on days 1-5 (QD×5) in a 21-day cycle[2]
- ARTISTRY-3 (NCT04592653) is a phase 1/2, open-label study evaluating less frequent IV (LFIV) dosing of nemvaleukin in advanced solid tumors
- With both QD×5 and LFIV regimens, nemvaleukin demonstrated expansion of CD8$^+$ T and natural killer (NK) cells in peripheral blood, with minimal expansion of regulatory T cells (T$_{regs}$). Similarly, administration of the nemvaleukin monotherapy QD×5 regimen in a patient with cutaneous melanoma increased granzyme B$^+$ (GZB$^+$) CD8$^+$ T cells, with minimal expansion of T$_{regs}$ in the tumor microenvironment (TME)[2-4]
- Here we report modulation of the TME in patients with advanced solid tumors, including ovarian cancer (OC) and mucosal melanoma (MM), dosed with 3 different schedules in ARTISTRY-3

Methods

Study Design and Key Eligibility Criteria

- ARTISTRY-3 is an ongoing phase 1/2, open-label study
- Escalating doses of nemvaleukin were evaluated across 3 schedules in 21-day cycles as follows: Schedule 1: dosing on day 1; Schedule 2: dosing on days 1 and 8; and Schedule 3: dosing on days 1 and 4 (**Figure 1**)
- Adult patients (≥18 years) with an Eastern Cooperative Oncology Group performance status (ECOG PS) of 0 or 1 and histologically/cytologically confirmed diagnosis of select malignant solid tumors having at least 1 qualifying target lesion (per Response Evaluation Criteria In Solid Tumors version 1.1 [RECIST v1.1]) were included
- The following key exclusion criteria were applied: active infection within 3 days of first scheduled dose for cycle 1, active autoimmune disease(s) requiring systemic treatment within the past 2 years, primary central nervous system malignancy, or prior IL-2- or IL-15-based therapy

Figure 1. ARTISTRY 3 Cohort 2 (Part A) study design

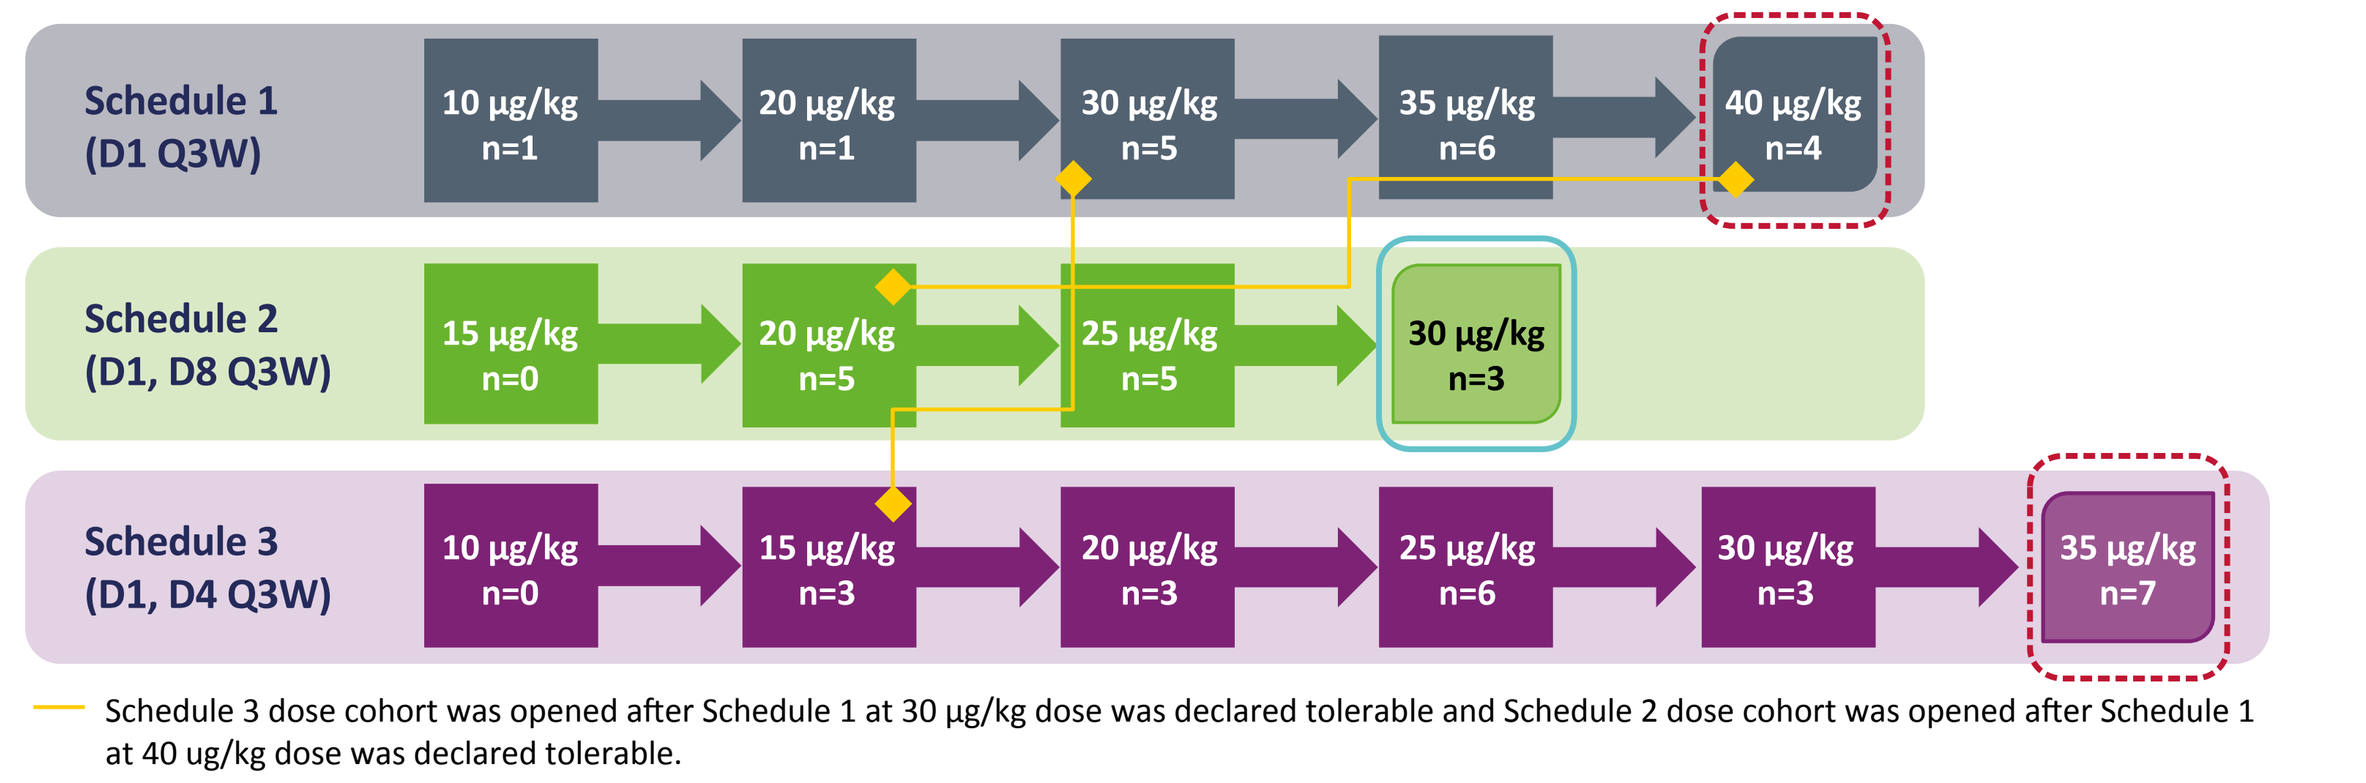

Schedule 1 (D1 Q3W)
10 µg/kg n=1
20 µg/kg n=1
30 µg/kg n=5
35 µg/kg n=6
40 µg/kg n=4
Schedule 2 (D1, D8 Q3W)
15 µg/kg n=0
20 µg/kg n=5
25 µg/kg n=5
30 µg/kg n=3
Schedule 3 (D1, D4 Q3W)
10 µg/kg n=0
15 µg/kg n=3
20 µg/kg n=3
25 µg/kg n=6
30 µg/kg n=3
35 µg/kg n=7
Schedule 3 dose cohort was opened after Schedule 1 at 30 µg/kg dose was declared tolerable and Schedule 2 dose cohort was opened after Schedule 1 at 40 ug/kg dose was declared tolerable.
Dose tolerable but not feasible in outpatient setting
Nemvaleukin LFIV RP2D

Dose escalation was based on Bayesian optimal interval design, with modifications to accommodate open enrollment and on predefined safety parameters of dose-limiting toxicity evaluated during the first cycle (21 days) by the safety review committee.
The maximum sample size was 30 patients per schedule. For all schedules, additional higher dose levels could continue in increments of 5 µg/kg from the highest dose (if tolerable and pharmacokinetic/pharmacodynamic parameters were not saturated) for at least 1 to 2 dose levels.
D, day; Q3W, every 3 weeks; RP2D, recommended phase 2 dose.

Assay

- Paired tumor biopsies were collected pre-treatment on screening day and while on treatment during cycle 2 day 8
- Deep immune profiling using a 14-marker panel encompassing various activation states of NK, CD8$^+$ T, CD4$^+$ T, and T$_{reg}$ cells, plasma cells, macrophages, and tumor markers was performed (**Table 1**)
- The MultiOmyx platform (NeoGenomics) was used to investigate the expression of 14 markers for cell quantification and spatial analytics, including summary density values of immune subsets in distinct tumor areas and proximity/neighborhood analysis[5]

Table 1. mIF panel

mIF panel	Cell type or function	mIF panel	Cell type or function
CD3	T cell	CD38	Activation
PD1	Activation/Exhaustion	NKp46	NK cell
CD4	CD4/Th	PanCK/SOX10	Tumor
CD163	Macrophage	FoxP3	T$_{reg}$
GZB	Cytolytic function	TCF-1/TCF-7	Stem-like progenitor
CD8	CD8/CTL	CD138	Plasma cell
CD45RO	Memory		

CD, cluster of differentiation; CTL, cytotoxic T cell; Fox, forkhead box; mIF, multiplex immunofluorescence; PanCK, pancytokeratin; PD1, programmed cell death protein 1; SOX, SRY-box transcription factor; TCF, T cell factor; Th, T helper.

Results

Baseline Patient Characteristics

- Paired biopsies were available from 8 patients across the 3 schedules
- Two of the 8 patients received 8 or more cycles of nemvaleukin treatment (**Table 2**)

Table 2. Baseline patient characteristics

Tumor type	ECOG PS	No. of prior anticancer therapies	Dosing schedule, dose group (µg/kg)	No. of treatment cycles received	Location of lesion biopsied (pre-treatment\|on-treatment)[a]	No. doses received, dose, days after last dose before on-treatment biopsy collection
MM (#1)[b]	1	5	1, 30	2	lymph node\|lymph node	2, 30 µg/kg, +7 days post dose
MM (#2)[b]	1	3	3, 15	2	lymph node\|pelvis	4, 15 µg/kg, +4 days post dose
MM (#3)[b]	1	6	3, 25	2	liver\|liver	3, 25 µg/kg[c], +3 days post dose
CM[b]	1	7	1, 30	2	adrenal gland\|adrenal gland	2, 30 µg/kg, +6 days post dose
OC (#1)[d,e]	0	3	2, 25	9	groin\|lymph node	4, 25 µg/kg, +3 days post dose
OC (#2)[d]	1	8	1, 30	8	spleen\|spleen	2, 30 µg/kg, +7 days post dose
OC (#3)[d]	1	14	3, 15	2	breast\|breast	4, 15 µg/kg, +6 days post dose
BC[f]	0	6	3, 35	2	breast\|breast	4, 35 µg/kg, +4 days post dose

[a]All pre-treatment biopsies were collected freshly within screening period except for that from 1 patient with OC (#3), whose pre-treatment biopsy was archival. [b]Prior therapies included anti-PD1- and anti-CTLA4-based immunotherapy, T-VEC, BRAF/MEK inhibitor, investigational agents, chemotherapy, and interferon alpha. [c]25 µg/kg at the first dose, then reduced to 20 µg/kg starting cycle 2 due to adverse event. [d]Prior therapies included platinum-/taxane-based and other cytotoxic chemotherapies, PARP inhibitors, antiangiogenics, hormonal therapies, and investigational agents. [e]Patient received prior pembrolizumab. [f]Prior therapies included hormonal therapy, CDK inhibitor, anti–programmed cell death ligand-1, investigational agent, antibody-drug conjugate, and chemotherapy.
BC, breast cancer; BRAF, B-raf proto-oncogene; CDK, cyclin-dependent kinase; CM, cutaneous melanoma; CTLA, cytotoxic T-lymphocyte–associated protein; MEK, mitogen-activated protein kinase; PARP, poly (adenosine diphosphate-ribose) polymerase; T-VEC, talimogene laherparepvec.

- Baseline infiltration of NK cells across tumor types was low (<100 cells/mm^2) (**Figure 2**)
 - Tumor-associated macrophages (TAMs) were the most abundant cell type across all tumors
 - Melanomas were generally more T cell inflamed than other tumor types, with the exception of that in 1 patient with OC (OC #2)

Figure 2. Patient-level immune cell densities in pre-treatment tumor biopsies

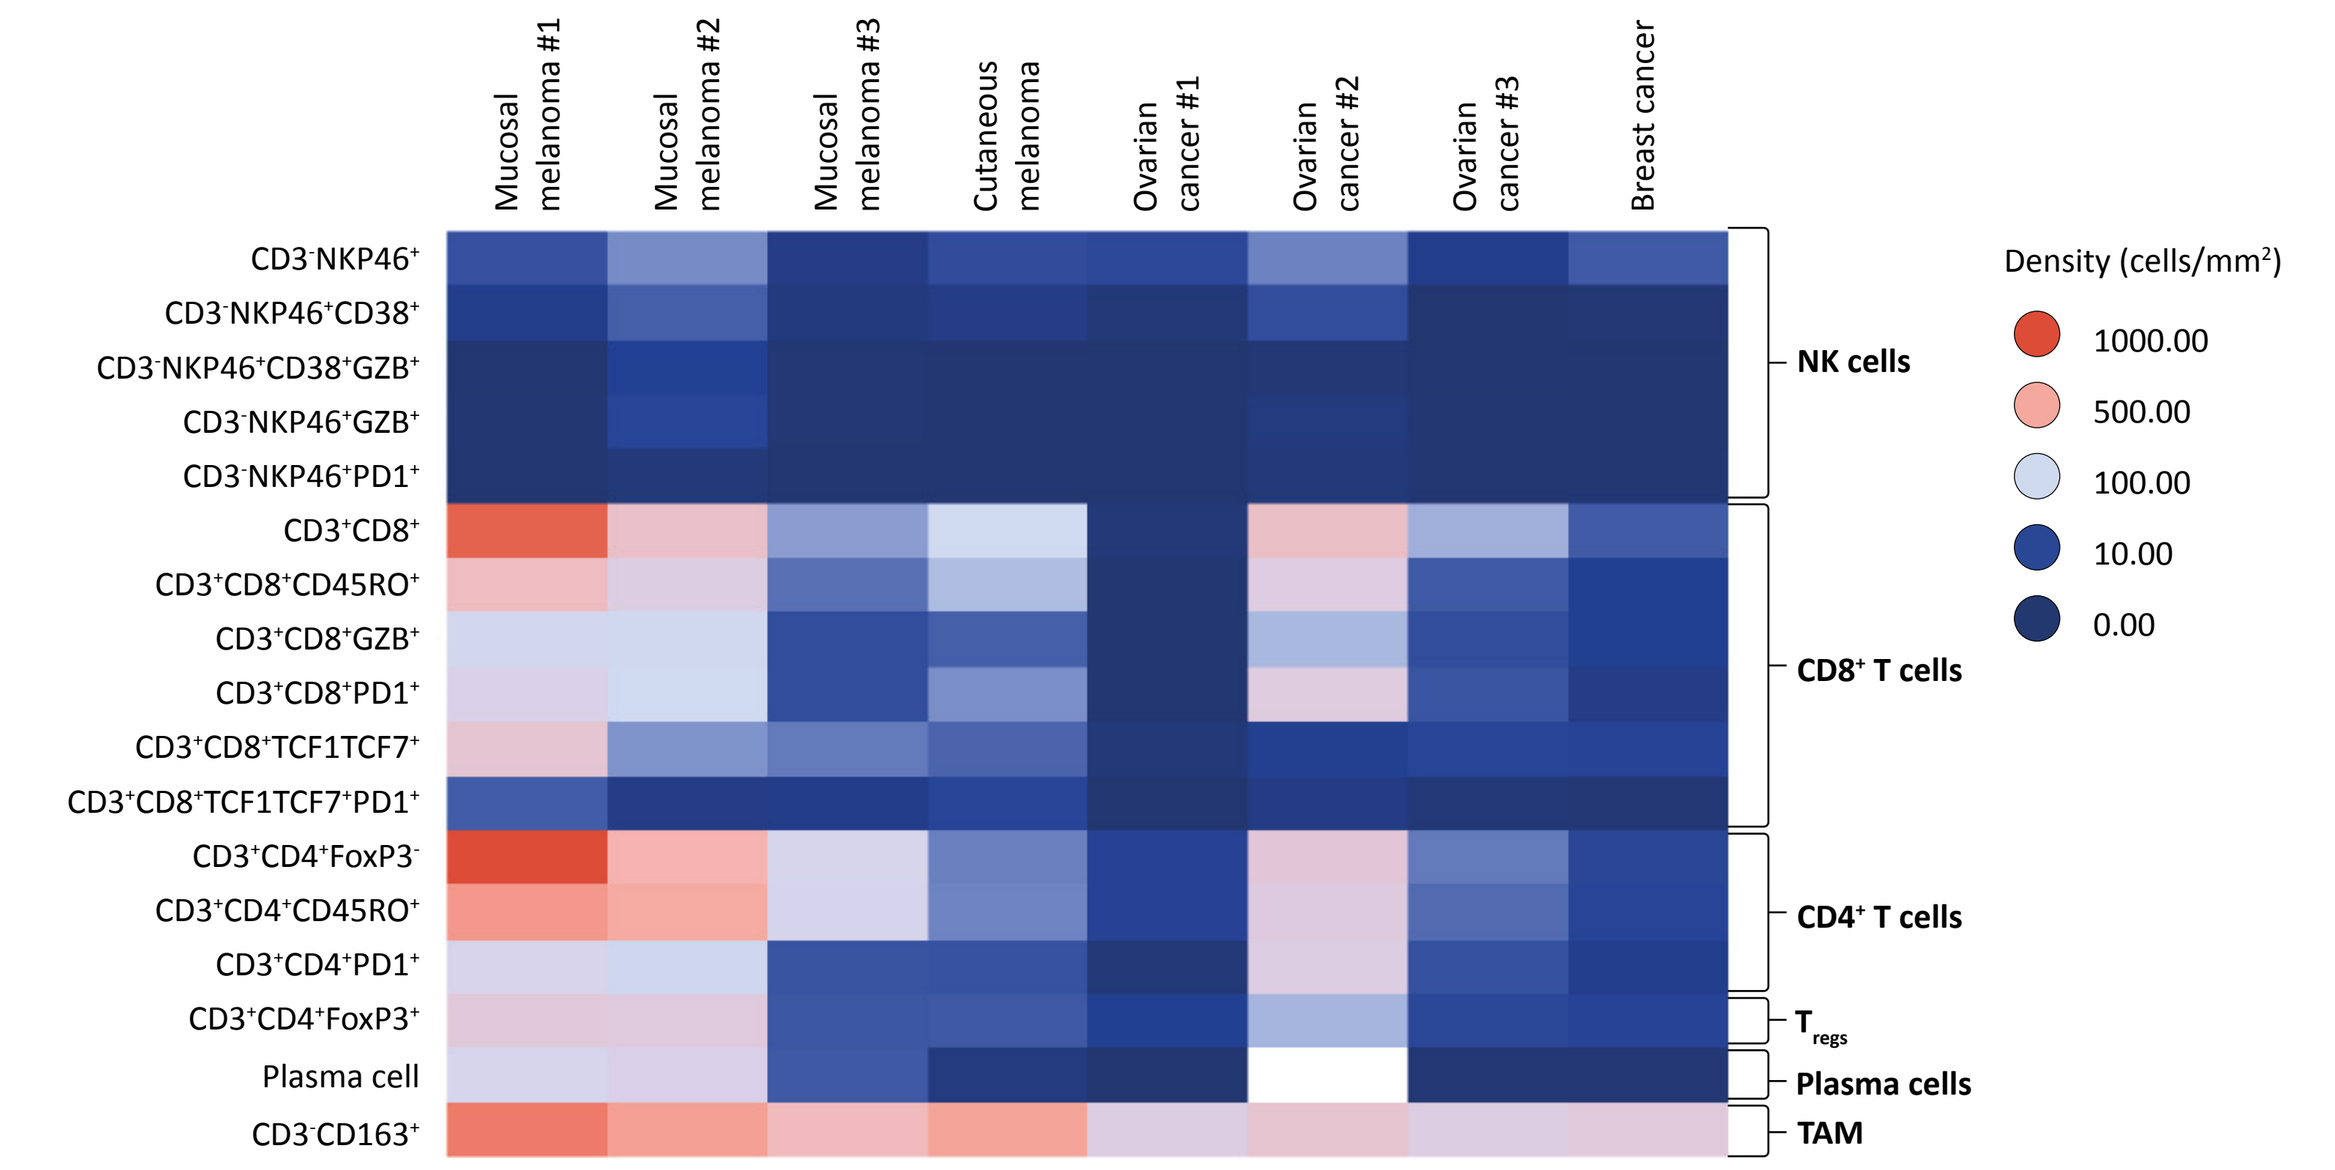

Mucosal melanoma #1
Mucosal melanoma #2
Mucosal melanoma #3
Cutaneous melanoma
Ovarian cancer #1
Ovarian cancer #2
Ovarian cancer #3
Breast cancer
CD3-NKP46+
CD3-NKP46+CD38+
CD3-NKP46+CD38+GZB+
CD3-NKP46+GZB+
CD3-NKP46+PD1+
CD3+CD8+
CD3+CD8+CD45RO+
CD3+CD8+GZB+
CD3+CD8+PD1+
CD3+CD8+TCF1TCF7+
CD3+CD8+TCF1TCF7+PD1+
CD3+CD4+FoxP3-
CD3+CD4+CD45RO+
CD3+CD4+PD1+
CD3+CD4+FoxP3+
Plasma cell
CD3-CD163+
NK cells
CD8+ T cells
CD4+ T cells
Tregs
Plasma cells
TAM
Density (cells/mm2)
1000.00
500.00
100.00
10.00
0.00

Whole tissue density outputs provided for 8 patients at baseline.

Changes in Leukocyte Population Densities Upon Nemvaleukin Treatment

- Increases in lymphoid populations, especially NK and CD8$^+$ T cells, were observed during nemvaleukin treatment. Increases in CD4$^+$ subsets and plasma cells were observed, except in MM tumors (**Figure 3**)
- On-treatment T$_{reg}$ densities varied, with some increases in density noted with concomitant effector expansion. However, these changes were minimal (**Figure 3**)
- Nemvaleukin treatment preferentially increased cytolytic effector NK and CD8$^+$ T cell densities compared with T$_{regs}$ in the TME (**Figure 3**)

Figure 3. Patient-level fold changes in immune populations and median densities of effector CD8$^+$ T and NK cells and T$_{regs}$ in on-treatment tumor biopsies

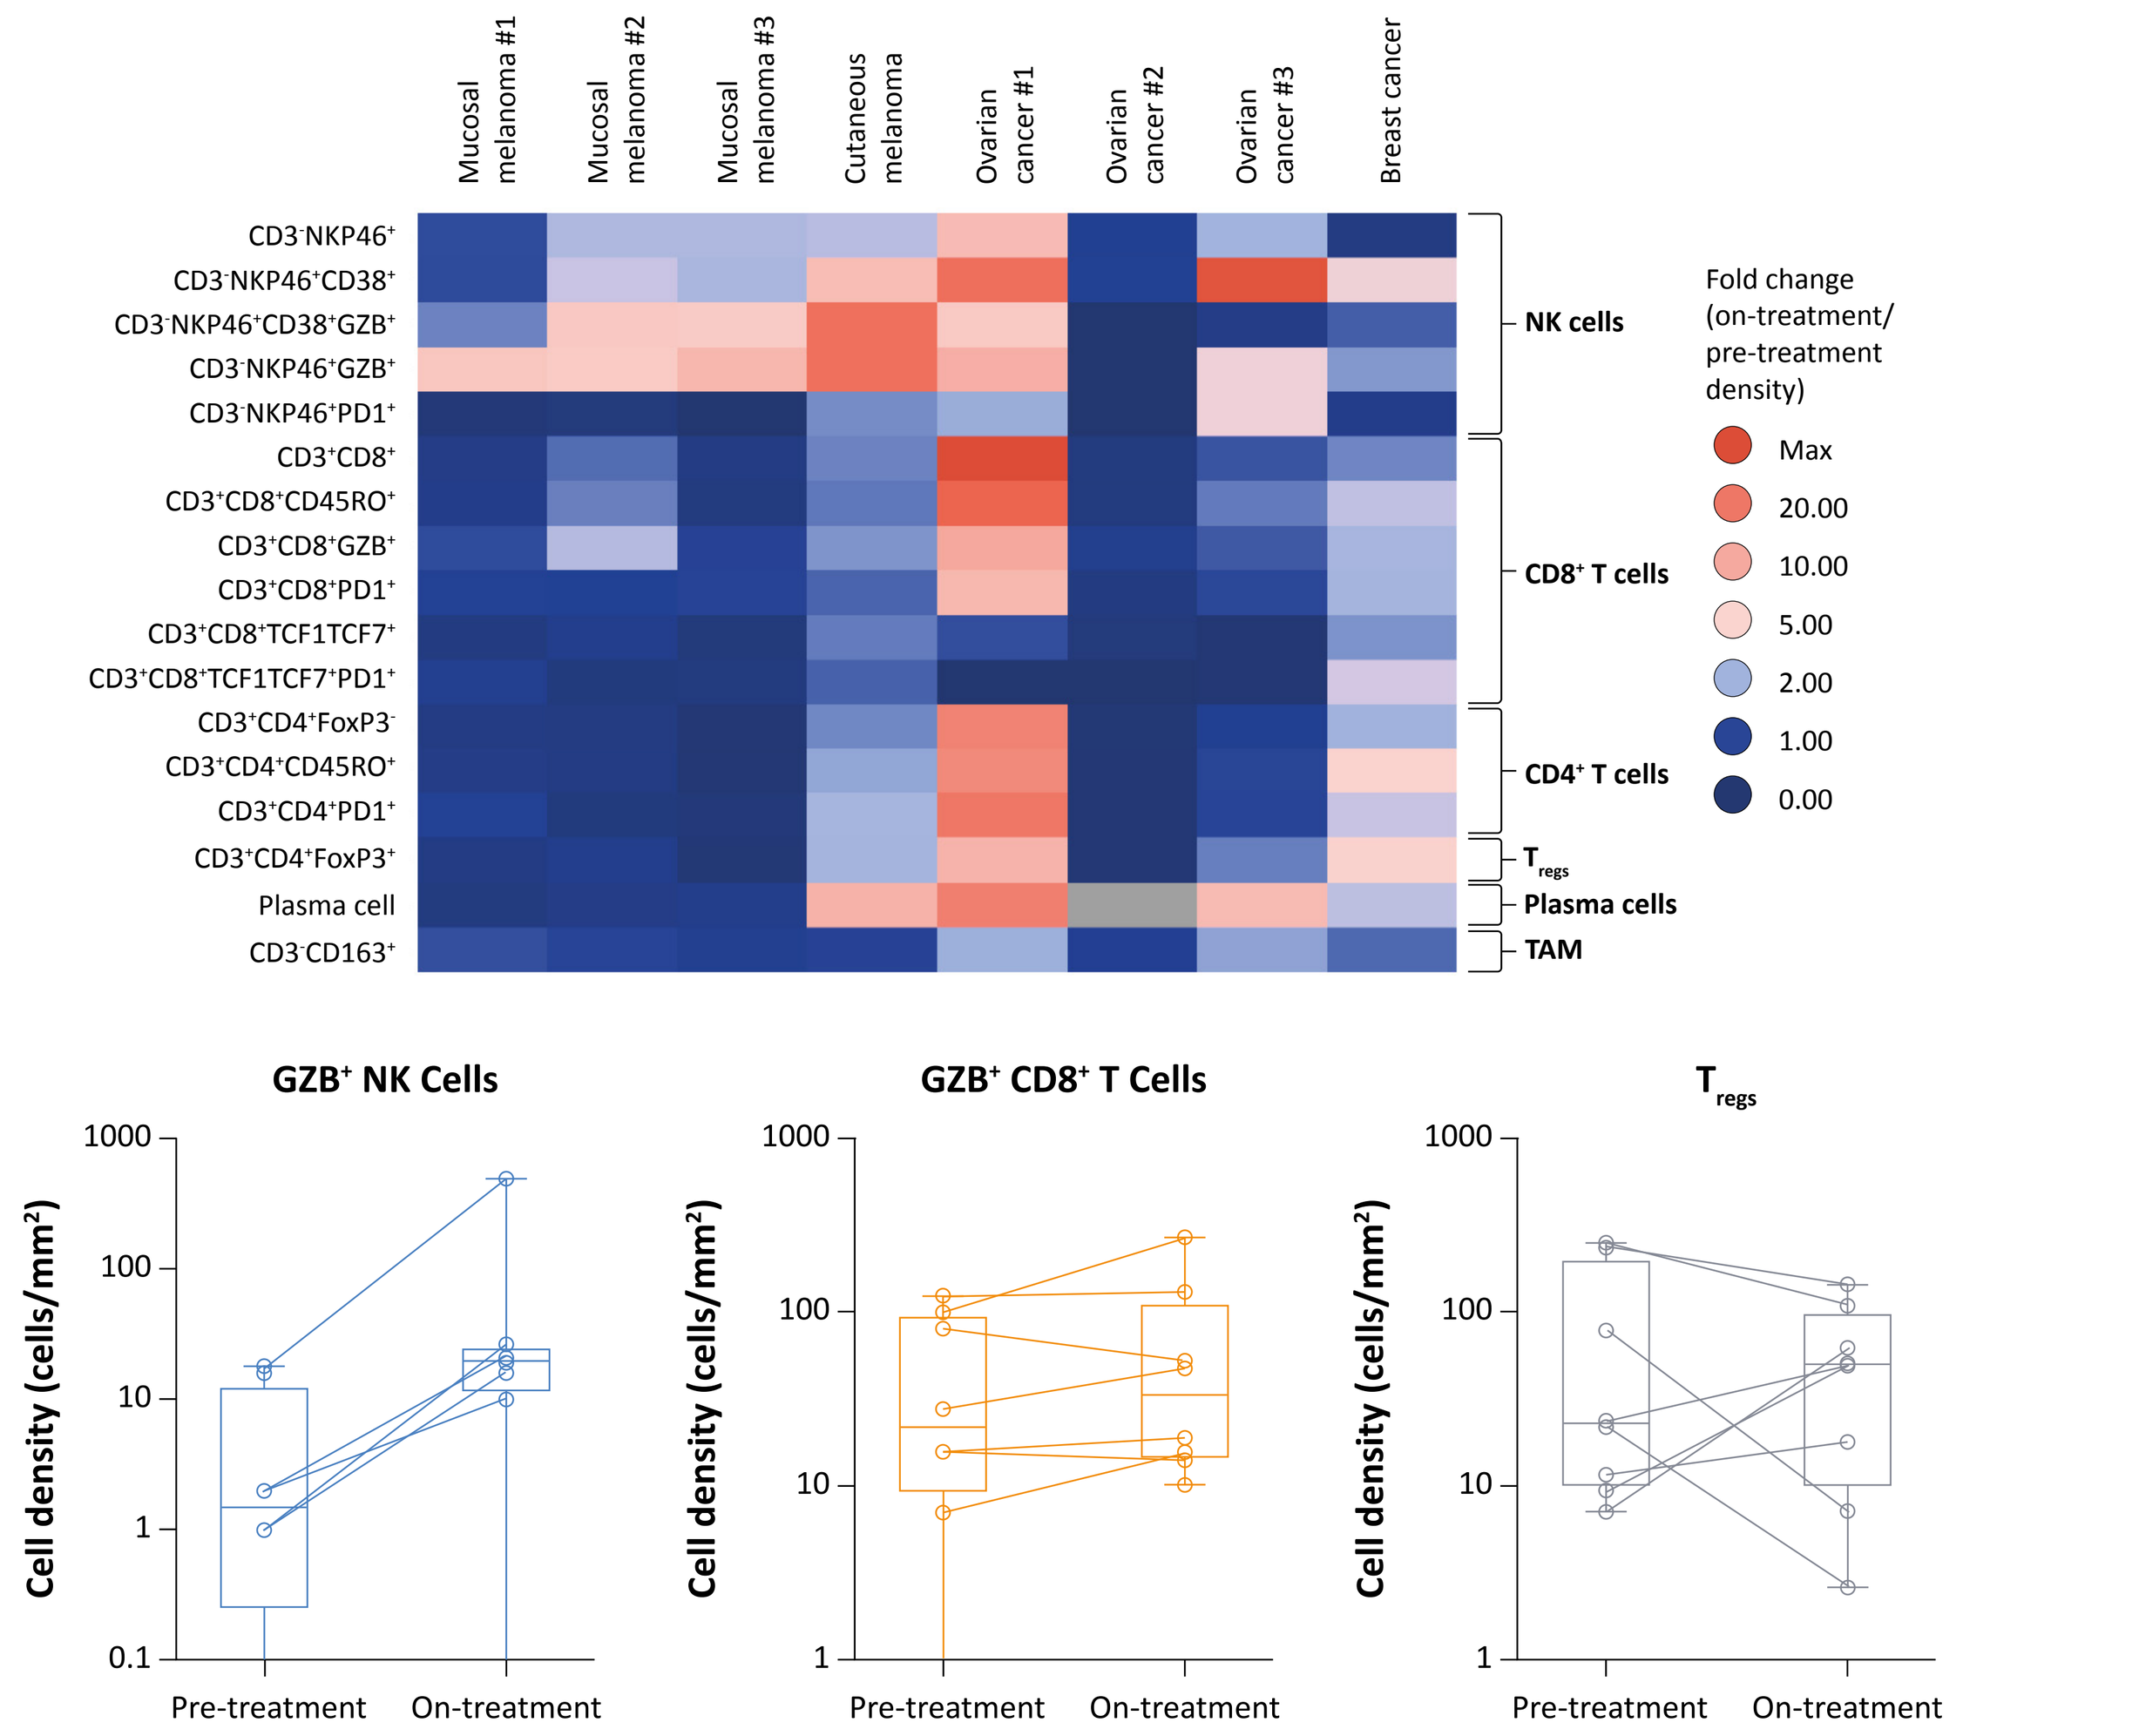

Mucosal melanoma #1
Mucosal melanoma #2
Mucosal melanoma #3
Cutaneous melanoma
Ovarian cancer #1
Ovarian cancer #2
Ovarian cancer #3
Breast cancer
NK cells
CD8+ T cells
CD4+ T cells
Tregs
Plasma cells
TAM
Fold change (on-treatment/ pre-treatment density)
Max
20.00
10.00
5.00
2.00
1.00
0.00
GZB+ NK Cells
GZB+ CD8+ T Cells
Tregs
Cell density (cells/mm2)
Pre-treatment
On-treatment

Whole tissue density outputs provided for 8 patients with paired pre-treatment and on-treatment biopsies. Before and after scatter of individual values overlaid with box and whisker plot. Box = IQR, line = median, whiskers = min/max. Zero values not plotted on log axis.
IQR, interquartile range.

- Increases in density ratios of NK:T$_{reg}$ and CD8$^+$:T$_{reg}$ were observed with less frequent nemvaleukin treatment, notably in MM #3 and OC #2 (**Figure 4**)

Figure 4. Effector immune cell to T$_{reg}$ ratios after nemvaleukin treatment in whole tissue paired biopsies

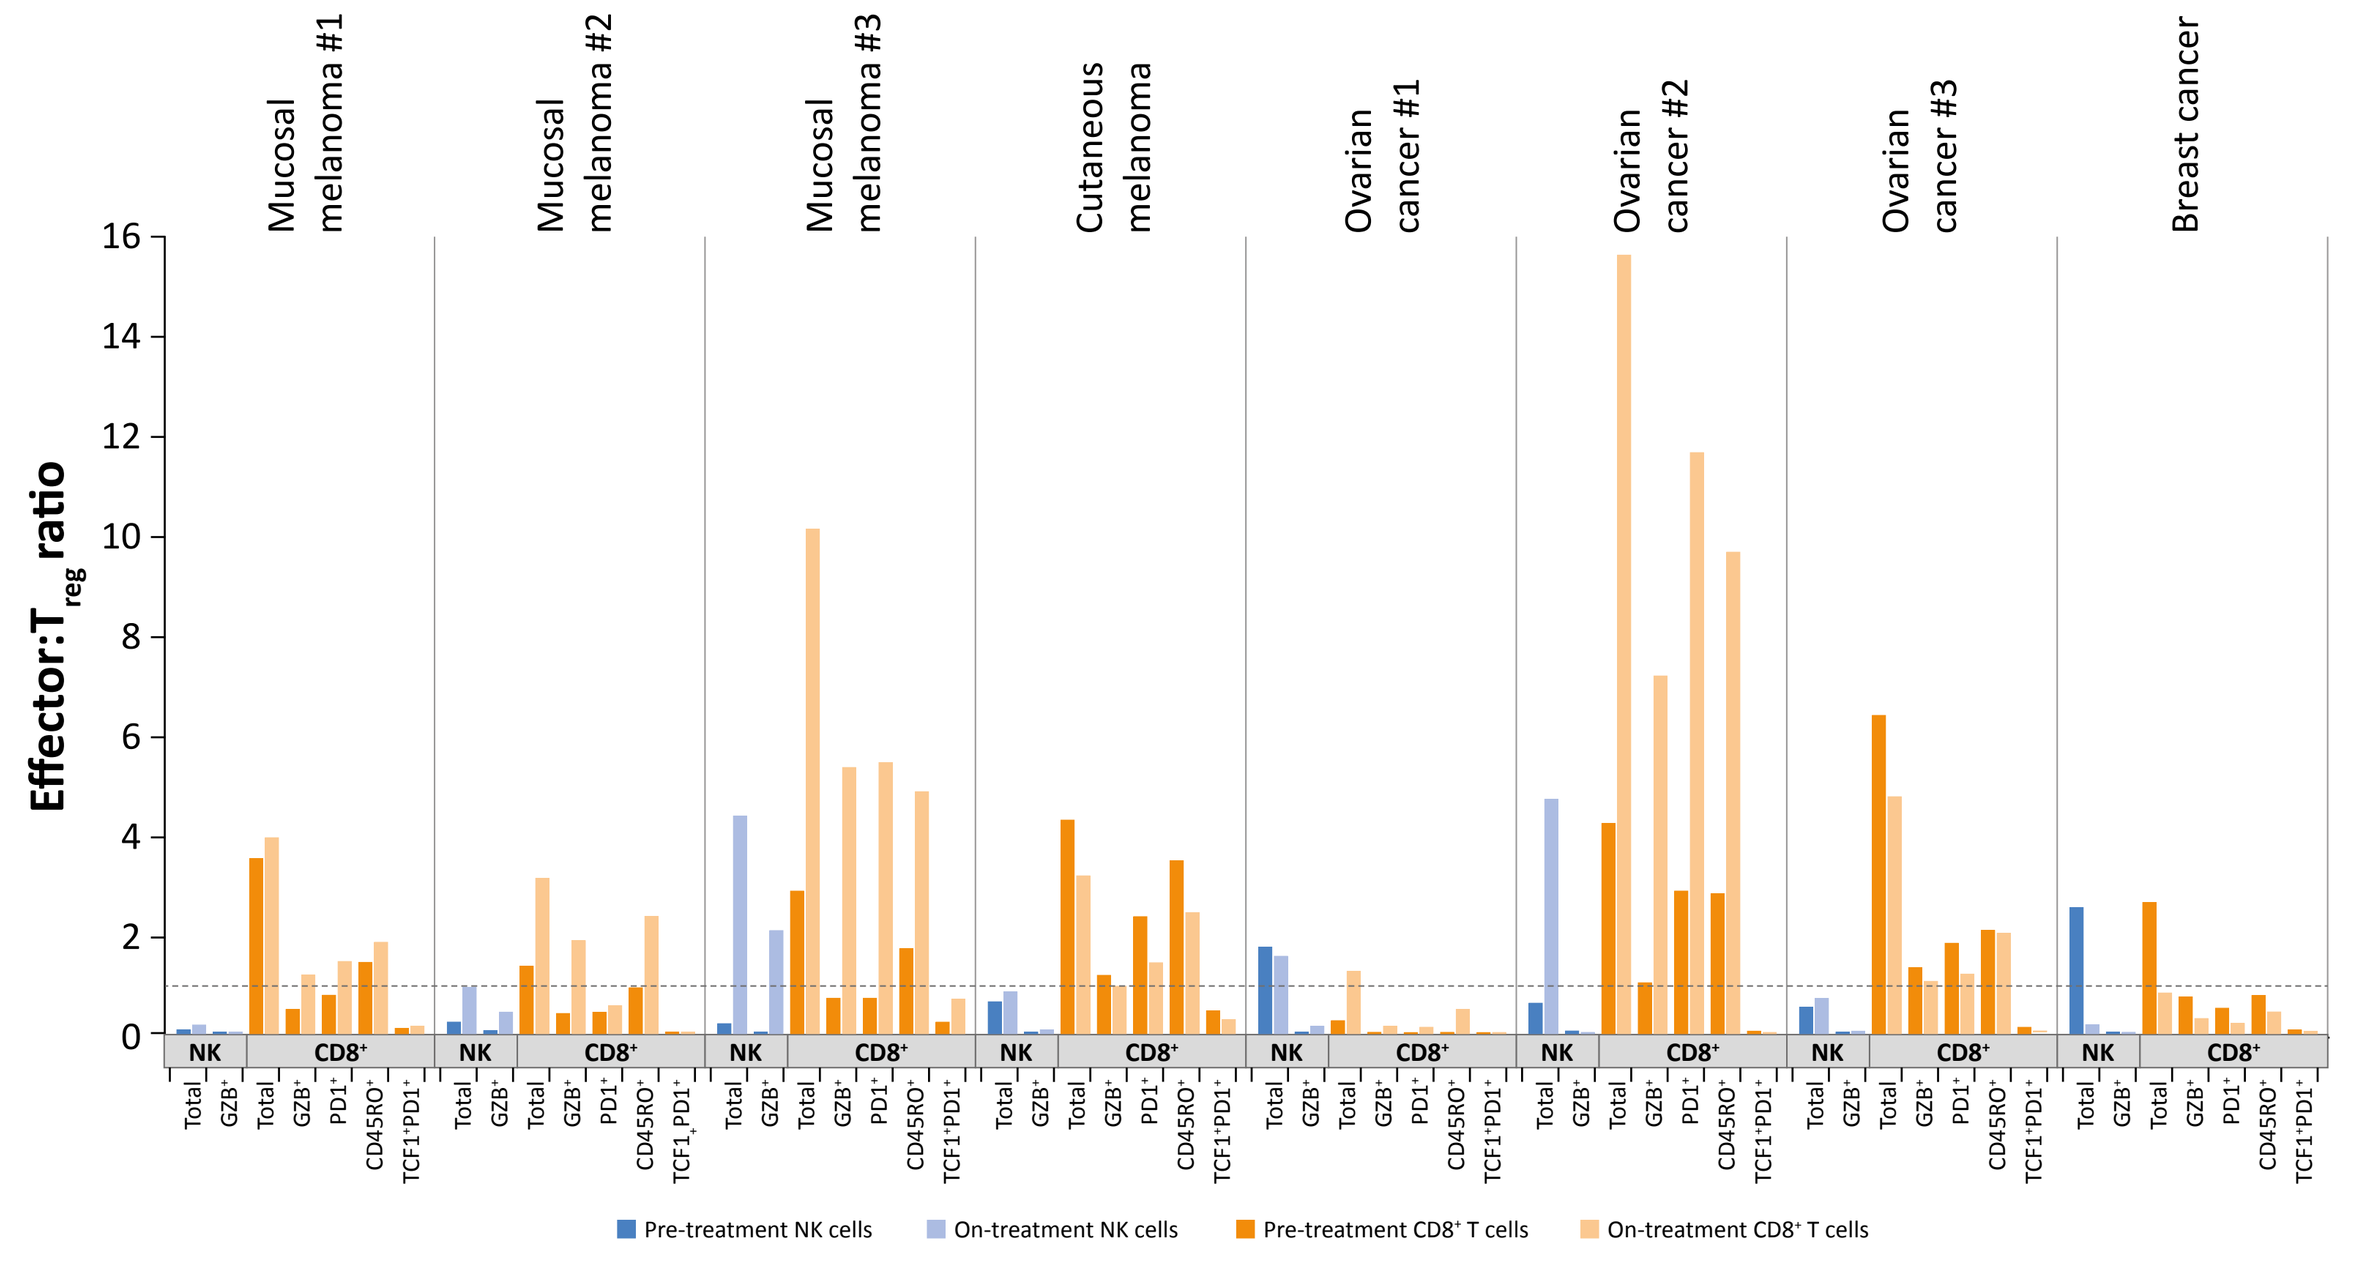

Mucosal melanoma #1
Mucosal melanoma #2
Mucosal melanoma #3
Cutaneous melanoma
Ovarian cancer #1
Ovarian cancer #2
Ovarian cancer #3
Breast cancer
Effector:Treg ratio
NK
CD8+
Total
GZB+
PD1+
CD45RO+
TCF1+PD1+
Pre-treatment NK cells
On-treatment NK cells
Pre-treatment CD8+ T cells
On-treatment CD8+ T cells

Whole tissue density outputs provided for 8 patients with paired pre-treatment and on-treatment biopsies. Dashed line represents ratio of 1.0.

Spatial Analytics

- Neighborhood analysis revealed increased tumor-proximal effector NK and CD8$^+$ T cells following nemvaleukin treatment in 7 of 8 patients; no pre-treatment values for spatial analytics were available for 1 patient (OC #2) (**Figure 5**)

Figure 5. Patient-level proximity analysis for GZB$^+$ NK and GZB$^+$ CD8$^+$ cells in tumor cell neighborhoods

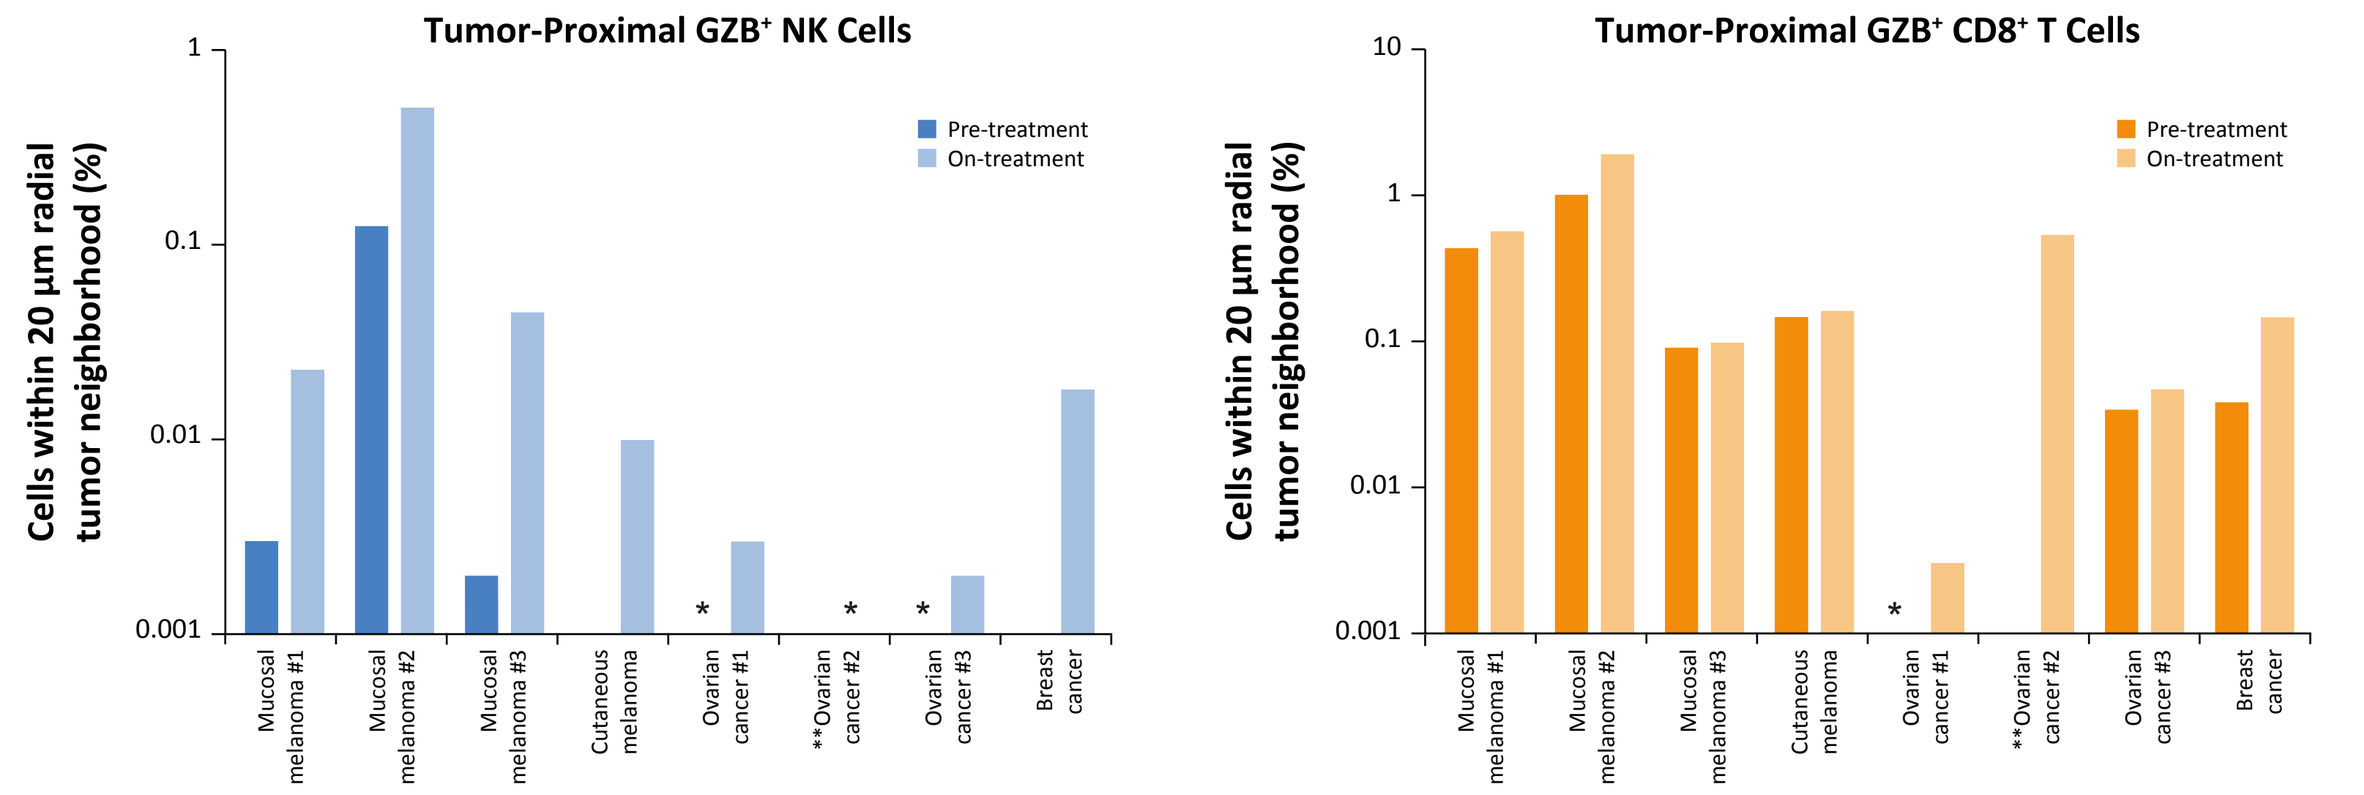

Tumor-Proximal GZB+ NK Cells
Tumor-Proximal GZB+ CD8+ T Cells
Cells within 20 µm radial tumor neighborhood (%)
Pre-treatment
On-treatment
Mucosal melanoma #1
Mucosal melanoma #2
Mucosal melanoma #3
Cutaneous melanoma
Ovarian cancer #1
**Ovarian cancer #2
Ovarian cancer #3
Breast cancer

*No target immune cells noted in tumor cell neighborhoods. **No screening value.

- Representative mIF images showing increased leukocyte populations in TME of on-treatment biopsies are depicted in **Figure 6**
- In the patient with MM (#2) (**Figure 6A**):
 - Pre-treatment biopsy was infiltrated with immune population rich in T$_{regs}$ and CD4$^+$ T cells, with few NK and CD8$^+$ T cells
 - In the on-treatment sample, induction of GZB$^+$ NK and CD8$^+$ T cells was observed
 - The results demonstrate a pharmacodynamic increase in cytolytic effectors, shifting the immune milieu from immunosuppressive to inflammatory
- In the patient with platinum-resistant OC (#1) (**Figure 6B**):
 - Pre-treatment biopsy was not immune infiltrated and exhibited a low density of lymphocytes
 - In the on-treatment sample, GZB$^+$ effector-rich lymphoid aggregate was observed, indicating immune activation
 - The patient exhibited a 20% decrease in target lesion and stable disease of >3 months, demonstrating some clinical benefit from induced immune populations

Figure 6. Representative H&E and mIF images of TME in paired biopsies

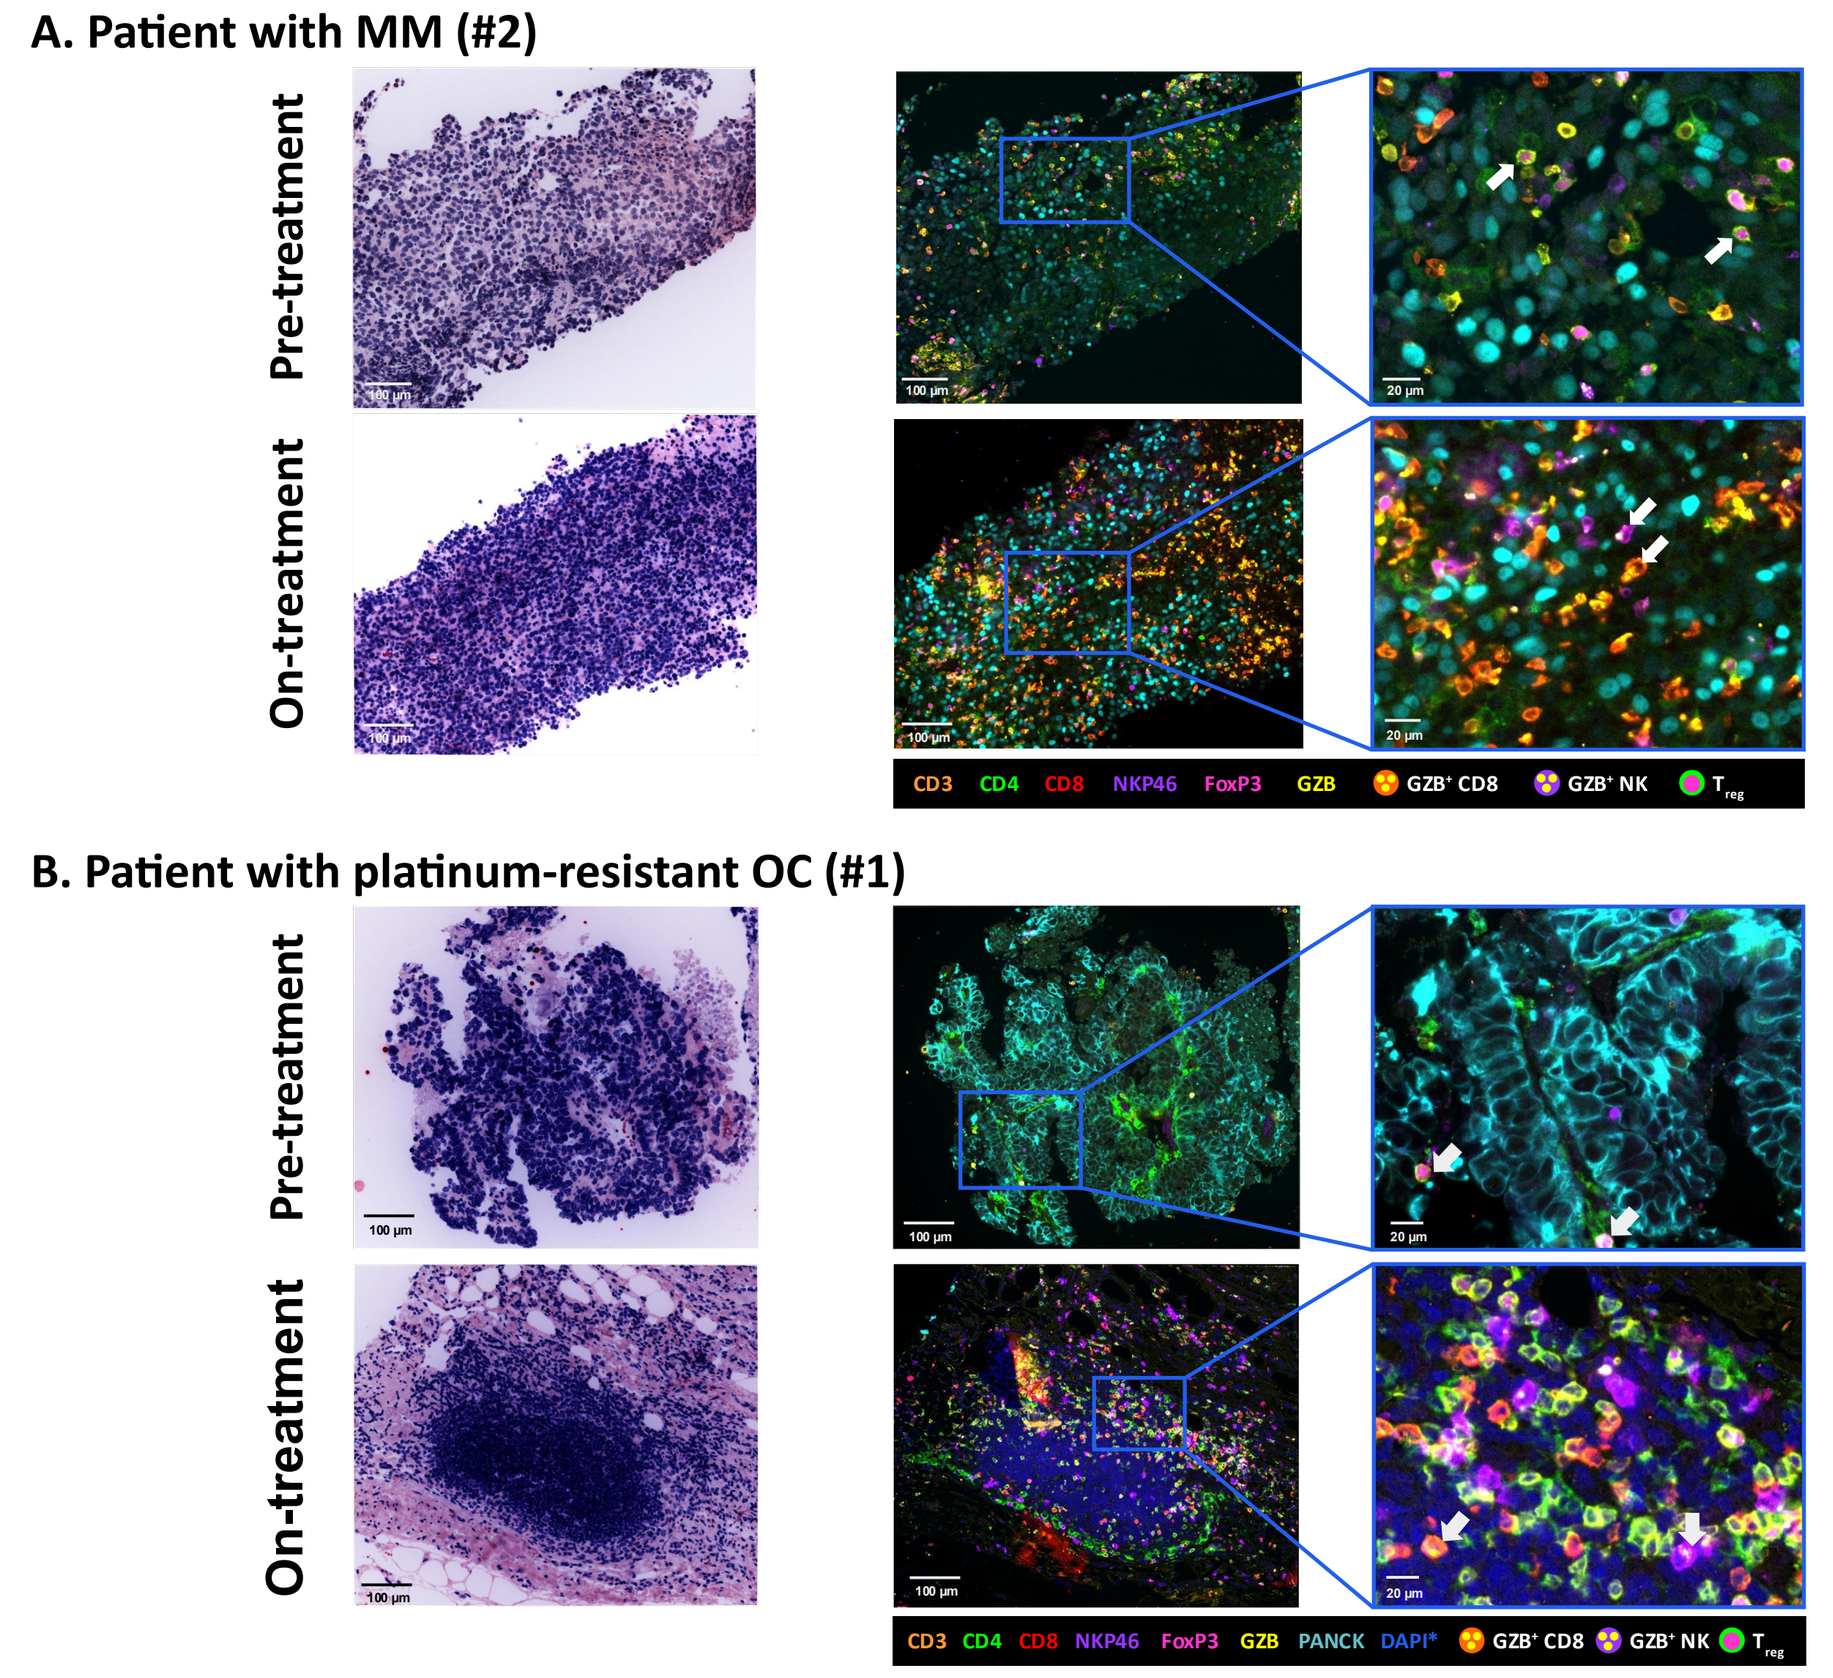

A. Patient with MM (#2)
Pre-treatment
On-treatment
CD3 CD4 CD8 NKP46 FoxP3 GZB GZB+ CD8 GZB+ NK Treg
B. Patient with platinum-resistant OC (#1)
Pre-treatment
On-treatment
CD3 CD4 CD8 NKP46 FoxP3 GZB PANCK DAPI* GZB+ CD8 GZB+ NK Treg

Magnification is 0.4× and 1.58×. White arrows denote T$_{regs}$ in pre-treatment biopsies, and GZB$^+$ NK and GZB$^+$ CD8 cells in on-treatment biopsies.
H&E, hemotoxylin and eosin.

References

1. Lopes JE, et al. *J Immunother Cancer*. 2020,8(1):e000673.
2. Vaishampayan U, et al. *J Clin Oncol*. 2022;40(16_suppl). Abstract #2500.
3. Piha-Paul SA, et al. *J Clin Oncol*. 2024;42(16_suppl). Abstract #2587.
4. Winer I, et al. *J ImmunoTher Cancer*. 2020;8.
5. Neo_Pharma_Multiplexed Immuoflourescence_Brochure_050422.pdf (neogenomics.com). Accessed August 28, 2024.
6. Lopes JE, et al. *J Pharmacol Exp Ther*. 2021;379:203-210.

Acknowledgments

- The authors would like to thank all the patients who are participating in this study and their families
- The study is sponsored by Mural Oncology
- Medical writing and editorial support was provided by Mandakini Singh, PhD, of Parexel and funded by Mural Oncology

Society for Immunotherapy of Cancer (SITC) 39th Annual Meeting, Houston, TX, USA • November 6-10, 2024