#432

Nemvaleukin Alfa, a Novel Engineered IL-2 Cytokine, in Combination With the Anti-PD-1 Antibody Pembrolizumab in Patients With Recurrent/Metastatic Head and Neck Squamous Cell Carcinoma (ION-01 Study)

Brian Gastman,[1] Martin A. Cheever,[2] Steven P. Fling,[2] Cesar A. Perez,[3] Manish R. Patel,[4] Jessica L. Geiger,[1] Zujun Li,[5] Marshall R. Posner,[6] Conor E. Steuer,[7] Leonard A. D'Amico,[2] Angela Shaulov Kask,[2] Yangchun Du,[8] Derek Matthies,[8] Sung Jin Huh,[8] Yan Wang,[8] Julie R. Graham,[8] Laura Q.M. Chow[9]

[1]Cleveland Clinic, Cleveland, OH; [2]Fred Hutchinson Cancer Research Center, Seattle, WA; [3]Sylvester Comprehensive Cancer Center, Miami, FL; [4]Masonic Cancer Center, Minneapolis, MN; [5]Laura and Isaac Perlmutter Cancer Center, New York, NY; [6]Icahn School of Medicine at Mount Sinai, New York, NY; [7]Winship Cancer Institute, Atlanta, GA; [8]Alkermes, Inc., Waltham, MA; [9]Dell Medical School, University of Texas at Austin, Austin, TX

INTRODUCTION

Nemvaleukin Alfa (Nemvaleukin) Is a Novel, Engineered Cytokine

- Selectively binds the intermediate-affinity IL-2R complex to preferentially activate CD8+ T cells and natural killer (NK) cells with minimal expansion of regulatory T cells (T_{regs}) (Figure 1).
- Designed to leverage proven antitumor effects of the IL-2 pathway while mitigating certain toxicities (Figure 1).

Figure 1: Cell Activation by IL-2 and Nemvaleukin

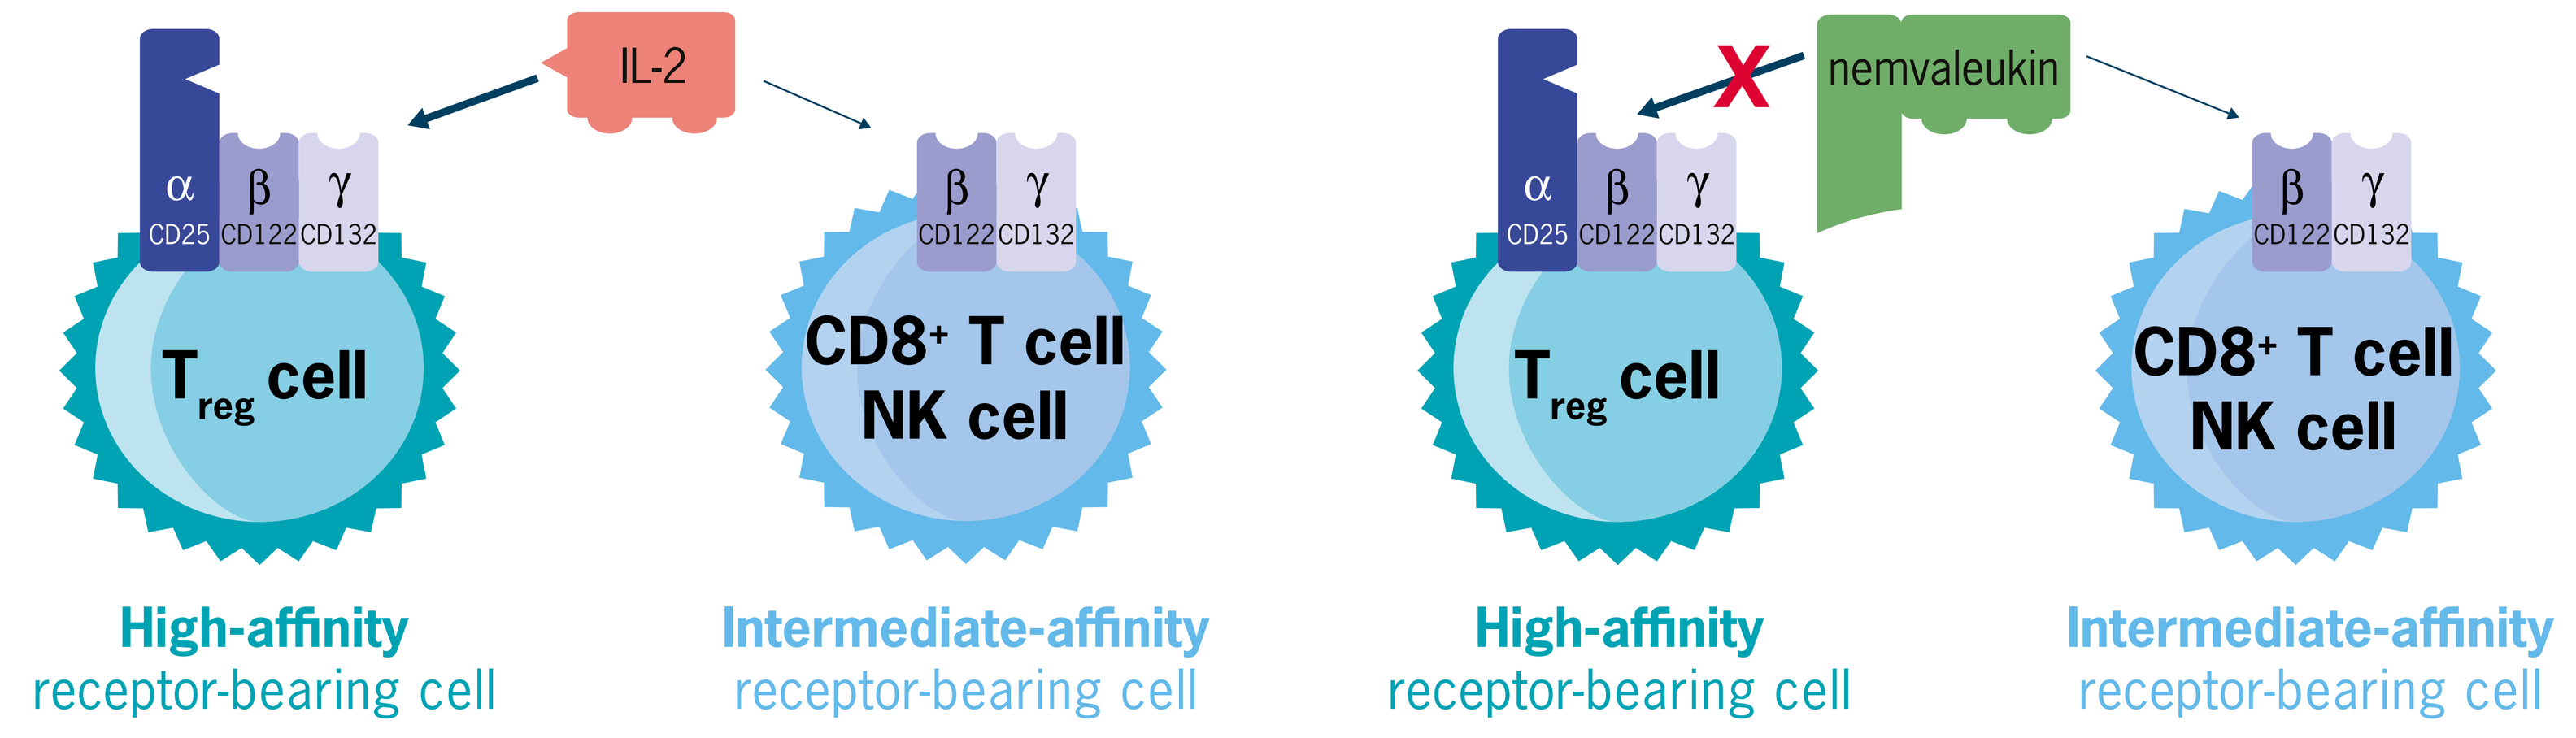
IL-2
α β γ
CD25 CD122 CD132
Treg cell
High-affinity
receptor-bearing cell
β γ
CD122 CD132
CD8+ T cell
NK cell
Intermediate-affinity
receptor-bearing cell
nemvaleukin
X
Treg cell
High-affinity
receptor-bearing cell
CD8+ T cell
NK cell
Intermediate-affinity
receptor-bearing cell

IL-2
- More potently activates the high-affinity IL-2R, which is preferentially expressed on immunosuppressive T_{regs}, is upregulated on vascular endothelial cells, and is associated with a high incidence of acute toxicities, including capillary leak syndrome.
- Preferential activation of high-affinity IL-2R-expressing cells by IL-2 leads to expansion of T_{regs}, which may counteract antitumor activity.

nemvaleukin
- Sterically occluded from binding to the high-affinity IL-2R.[1]
- Designed to selectively bind to the intermediate-affinity IL-2R for preferential activation and expansion of tumor-killing CD8+ T cells and NK cells, with minimal activation of T_{regs}, as well as mitigate toxicities associated with the preferential binding of IL-2 to high-affinity IL-2R.[1]

Clinical and Preclinical Studies Support the Potential for the Broad Use of Nemvaleukin

- Antitumor activity of multiple agents was enhanced when each was used in combination with nemvaleukin in preclinical studies.
- Responses with pembrolizumab were enhanced when it was combined with nemvaleukin in a variety of tumor types, including platinum-resistant ovarian, breast, cervical, lung, head and neck, gastrointestinal, and genitourinary.[2,3]
- Antitumor activity with nemvaleukin monotherapy was seen in patients who were previously treated with and progressed on checkpoint inhibitor (CPI) therapy in melanoma and renal cell carcinoma.[4] These results led to Orphan Drug and Fast Track designations of nemvaleukin for treatment of mucosal melanoma by the United States Food and Drug Administration.
- Both intravenous (IV) and subcutaneous (SC) administrations of nemvaleukin demonstrated dose-dependent, selective expansion of CD8+ T and NK cells, with minimal expansion of T_{regs}.[4,5]

METHODS

- ION-01 (NCT04144517) is a nonrandomized trial in adult patients with histologically or cytopathologically confirmed diagnosis of metastatic or recurrent head and neck squamous cell carcinoma.
- Eligible patients in the 2 cohorts reported here most recently received anti-PD-(L)1 therapy, and their disease progressed on this therapy.
- The primary endpoint is the rate of new or improved antitumor response after the addition of nemvaleukin.
- Secondary objectives include characterization of the antitumor response and evaluation of safety and tolerability of the combination regimen.
- Patients receive IV nemvaleukin (3 μg/kg) once daily for the first 5 days and pembrolizumab (200 mg) on day 1 of each 21-day cycle.
- Tumor imaging was performed after every second cycle.
- Pharmacodynamic activity and immune cell subpopulations were assessed in whole blood using 21-color flow cytometry.
- We present pharmacodynamic, safety, and antitumor activity (RECIST v1.1) data as of the data cutoff date of July 2021.

RESULTS

Patient Population

- 14 patients with progressive disease to anti-PD-(L)1 therapy received combination therapy with nemvaleukin and pembrolizumab.
 - 8 had previous best response of progressive disease and 6 had previous best response of stable disease or partial response to anti-PD-(L)1 therapy.
 - Median age was 64.5 years, 86% were male, and all were Caucasian.
 - Prior anticancer therapy included systemic therapy (100%), radiotherapy (93%), and surgery (50%).
 - Eastern Cooperative Oncology Group performance status was 0 (14%) and 1 (86%).

Pharmacodynamics

- Total NK and CD8+ T cell populations increased with combination therapy (3.5-fold and 1.7-fold, respectively; Figure 2A).
 - The proportion of $CD56_{Bright}$ NK cells increased post treatment (Figure 2B).
 - The total CD8+ T cell increase was primarily due to expansion of effector and terminally differentiated effector CD8+ T cells (Figure 2C).

Figure 2: NK Cells and CD8+ T Cells With Nemvaleukin and Pembrolizumab Combination Therapy

A. Maximum Fold Change

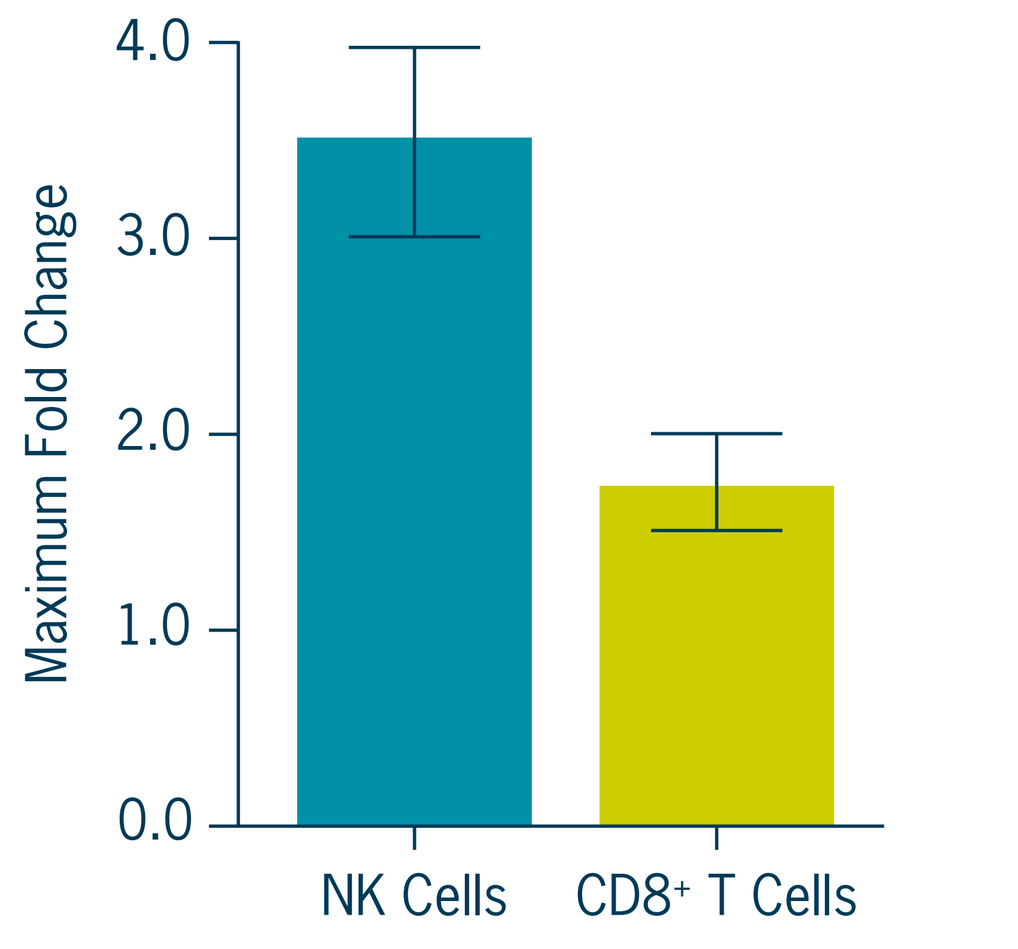
Maximum Fold Change
4.0
3.0
2.0
1.0
0.0
NK Cells
CD8+ T Cells

B. Proportion of NK Cell Subtypes

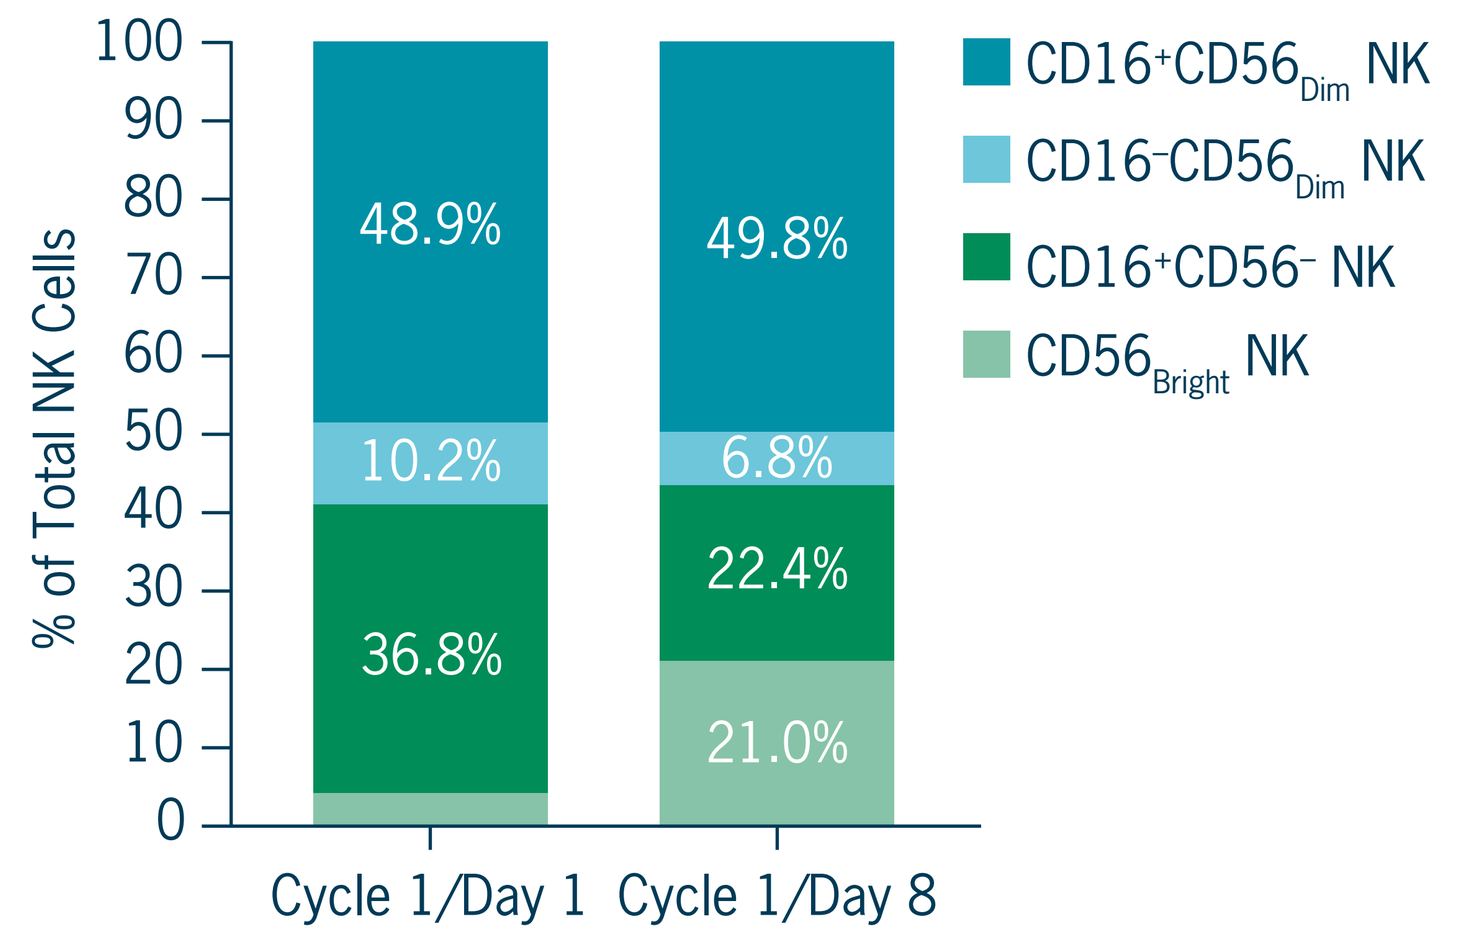
% of Total NK Cells
100
90
80
70
60
50
40
30
20
10
0
48.9%
10.2%
36.8%
49.8%
6.8%
22.4%
21.0%
Cycle 1/Day 1
Cycle 1/Day 8
CD16+CD56Dim NK
CD16-CD56Dim NK
CD16+CD56- NK
CD56Bright NK

C. Absolute Counts of CD8+ T Cell Subtypes

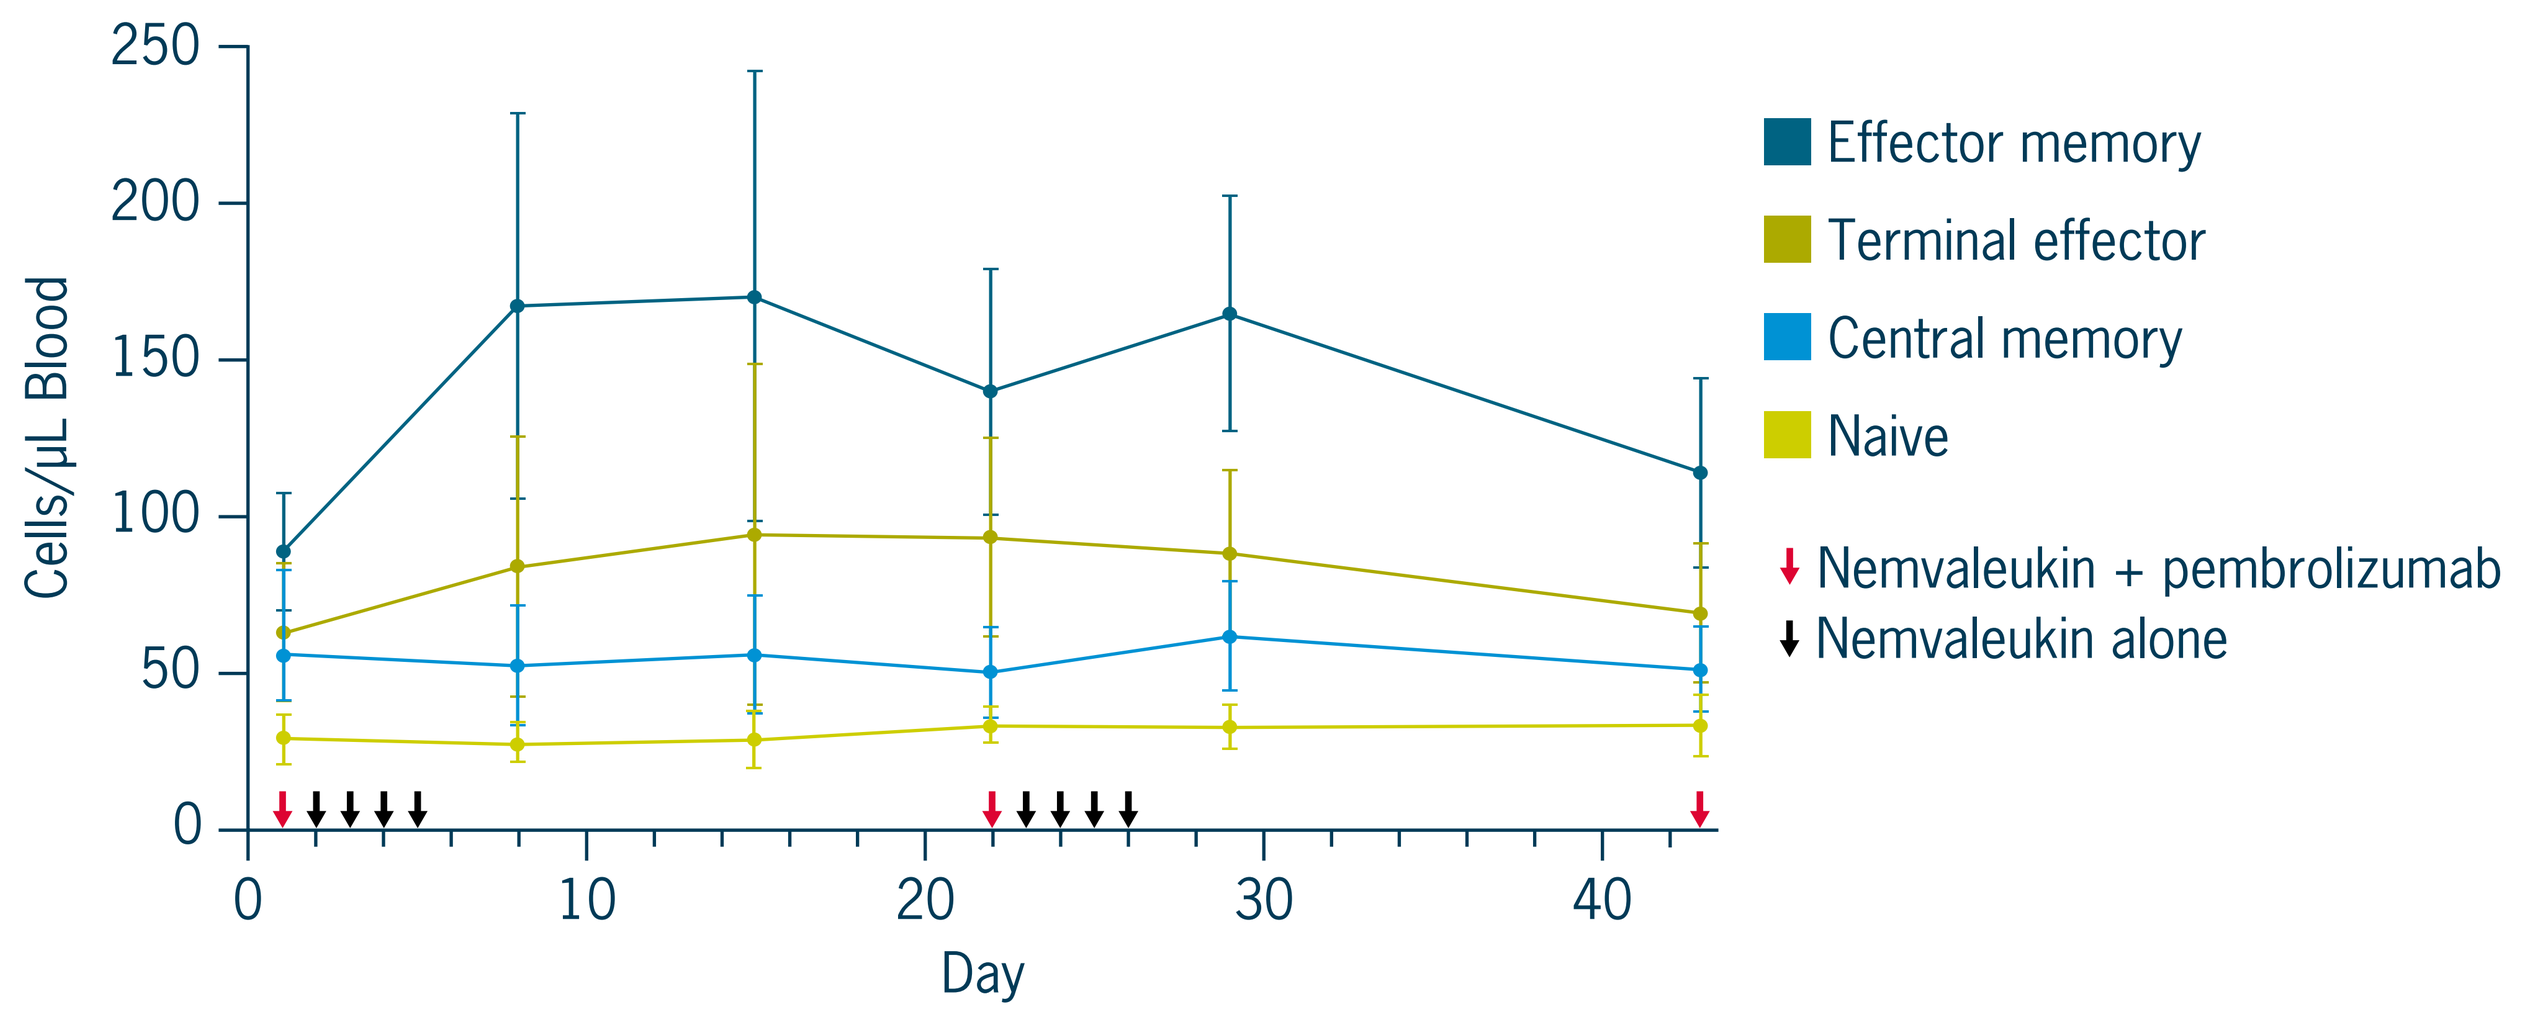
Cells/μL Blood
250
200
150
100
50
0
0
10
20
30
40
Day
Effector memory
Terminal effector
Central memory
Naive
Nemvaleukin + pembrolizumab
Nemvaleukin alone

Safety

- Overall, among the 14 patients who received treatment, the nemvaleukin safety profile was consistent with that reported for IV nemvaleukin in other clinical studies, with no new signals observed (Table 1).
- One patient discontinued treatment due to nemvaleukin-related grade 3 worsening hypertension and chills, grade 2 tachycardia, cytokine release syndrome, and fatigue, and grade 1 dyspnea.
- There were no treatment-emergent events leading to dose reduction or death.
- Relative to safety results in other clinical trials of nemvaleukin monotherapy, no additive toxicity was observed with the addition of pembrolizumab to the treatment regimen.

Antitumor Activity

- Responses and duration of response are shown in Figure 3.
- One patient achieved a partial response (Figure 3) with complete resolution in the target lesion (Figure 4), and has completed 1 year of treatment per protocol, following the data cutoff date.

Table 1: Safety Summary

Event, n (%)	N = 14		
AE summary			
Any AE, regardless of causality	14 (100.0)		
Grade 1 or 2 nemvaleukin-related AE	7 (50.0)		
Grade 3 or 4 nemvaleukin-related AE	4 (28.6)		
AEs, regardless of causality, in ≥30% of patients overall	**Grade 1/2**	**Grade 3**	**Grade 4**
Chills	8 (57.1)	1 (7.1)	0
Pyrexia	7 (50.0)	2 (14.3)	0
Fatigue	8 (57.1)	0	0
Hypoalbuminemia	6 (42.9)	0	0
Hypotension	4 (28.6)	1 (7.1)	0
Nausea	5 (35.7)	0	0
Nemvaleukin-related AEs in ≥30% of patients overall	**Grade 1/2**	**Grade 3**	**Grade 4**
Chills	8 (57.1)	1 (7.1)	0
Pyrexia	6 (42.9)	2 (14.3)	0
Fatigue	6 (42.9)	0	0
Nausea	5 (35.7)	0	0

Figure 3: Duration of Treatment and Summary of Responses

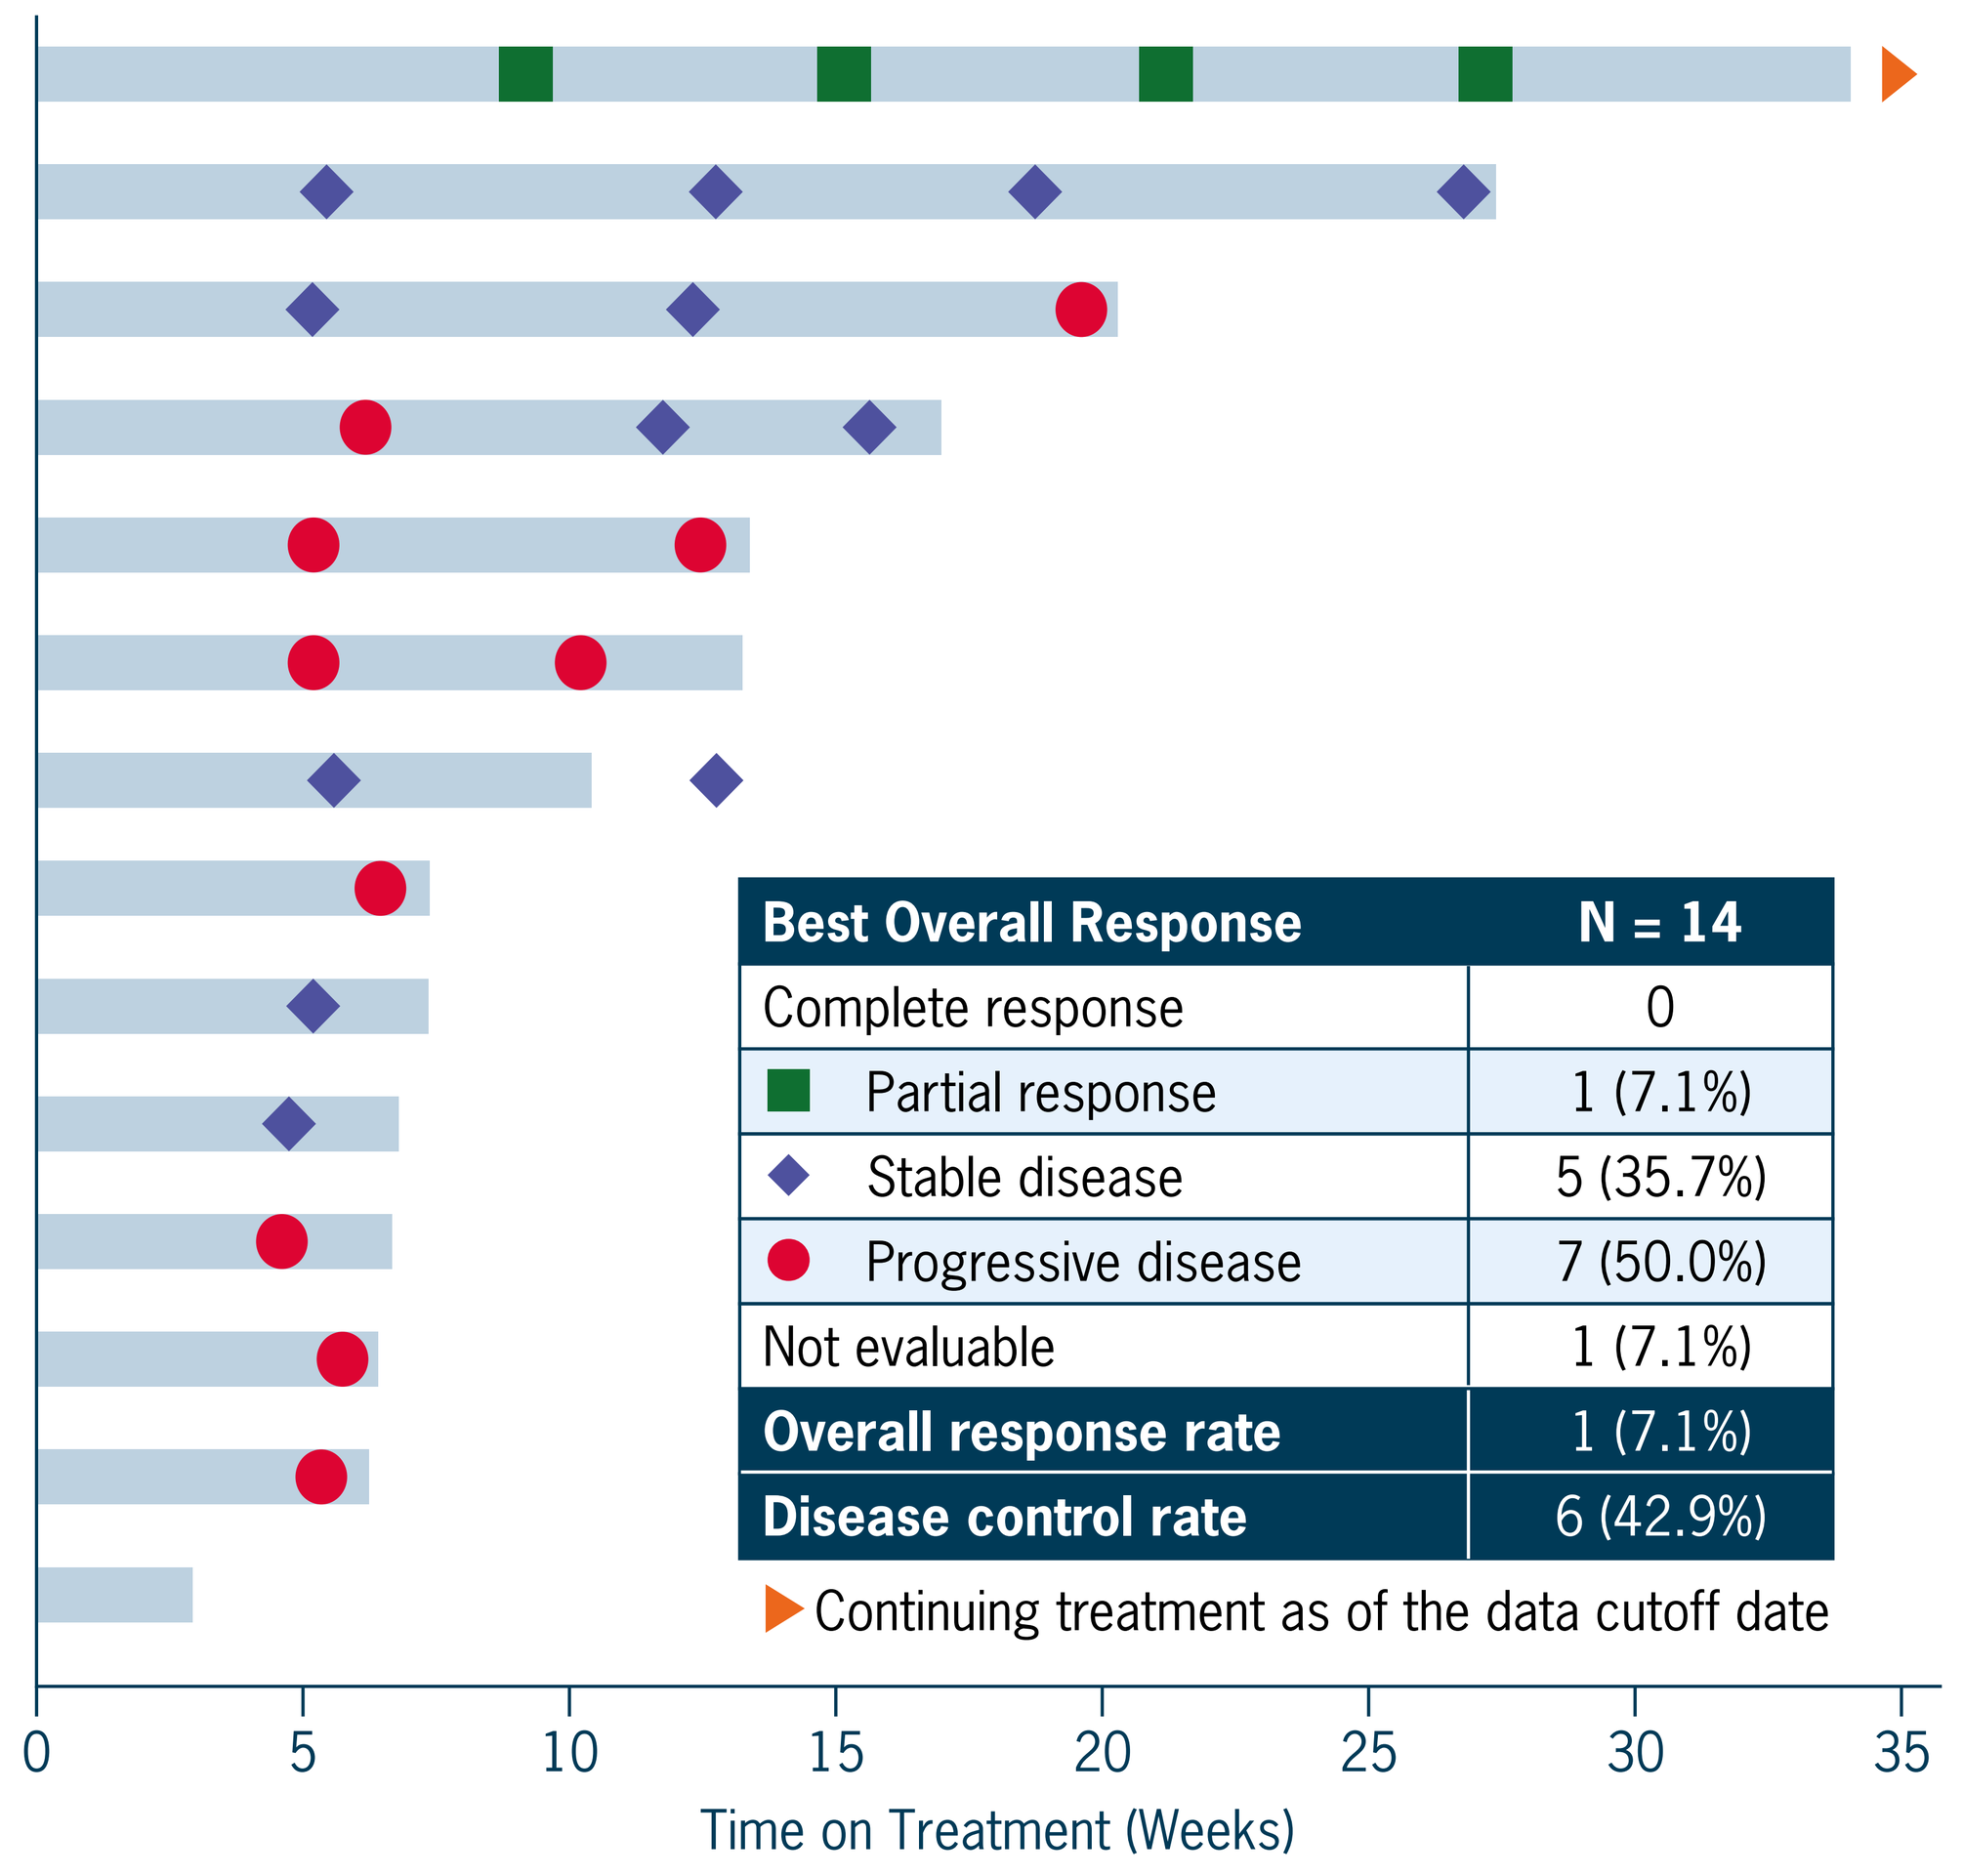
0
5
10
15
20
25
30
35
Time on Treatment (Weeks)

Best Overall Response	N = 14
Complete response	0
Partial response	1 (7.1%)
Stable disease	5 (35.7%)
Progressive disease	7 (50.0%)
Not evaluable	1 (7.1%)
Overall response rate	1 (7.1%)
Disease control rate	6 (42.9%)

Continuing treatment as of the data cutoff date

Figure 4. Durable Complete Resolution of Target Lesion in Soft Tissue Component Adjacent to Right Fourth Rib With Nemvaleukin and Pembrolizumab Combination Therapy

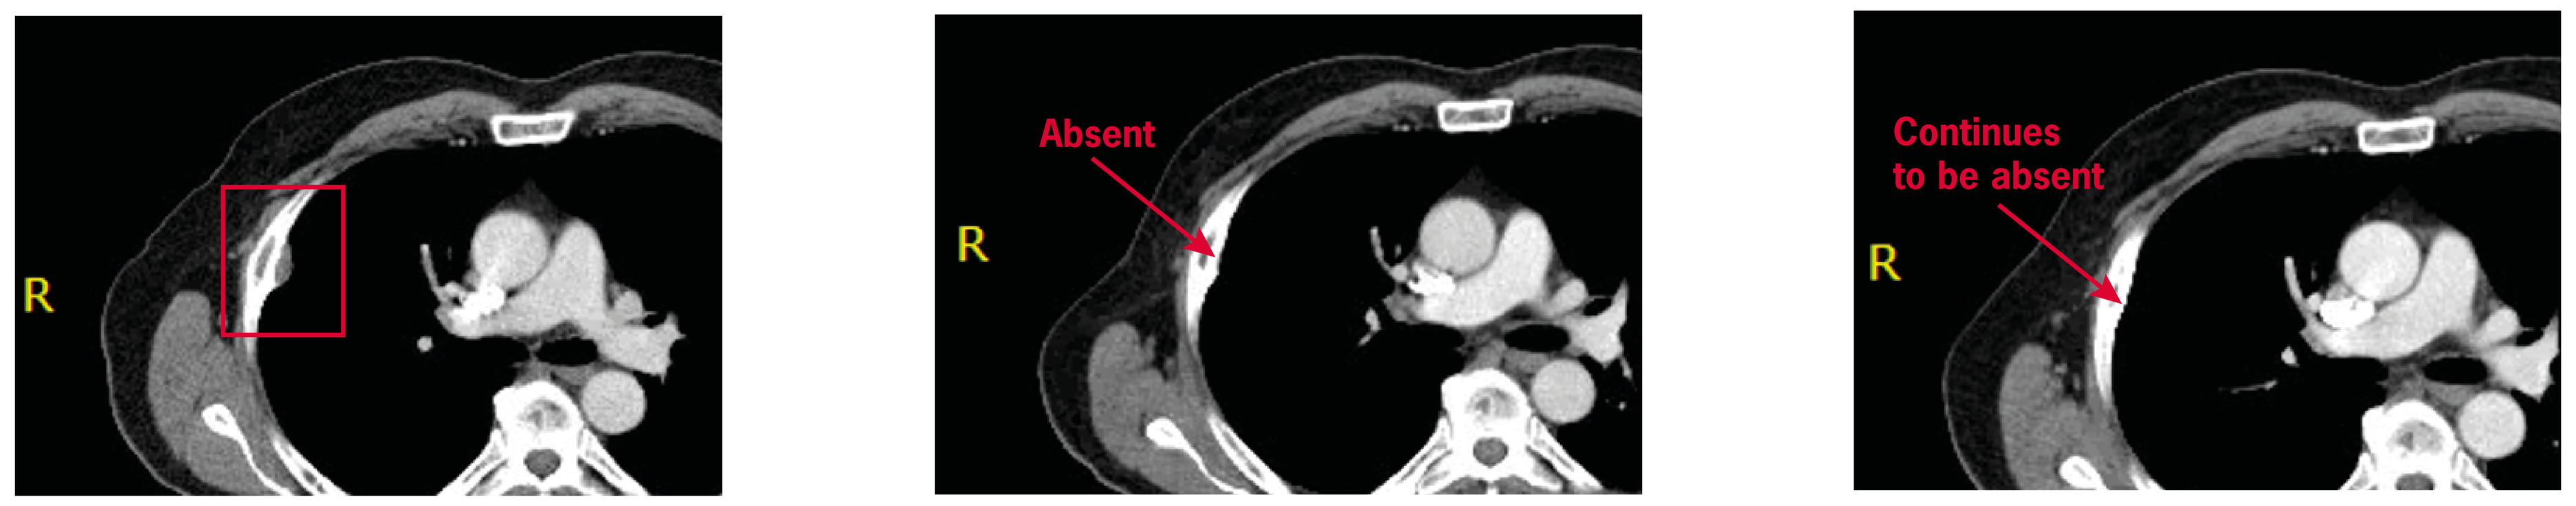
R
Absent
R
Continues
to be absent
R

Baseline: 10/15/2020 | **Pre-cycle 3: 01/04/2021** | **Pre-cycle 13: 08/16/2021[a]**

[a]After the data cutoff date.

CONCLUSIONS

- Nemvaleukin and pembrolizumab combination therapy was generally well tolerated; adverse events were consistent with those observed with IV nemvaleukin monotherapy in ARTISTRY studies.[4]
- Peripheral immune cell expansion profiles are comparable to those observed with the same combination regimen in the ARTISTRY-1 phase 1 study.
- These data warrant further evaluation of nemvaleukin.
 - Head and neck squamous cell carcinoma and other solid tumors are included in the ARTISTRY-2 phase 1/2 study (NCT03861793) of SC nemvaleukin as monotherapy and in combination with pembrolizumab.
 - A phase 3 study of nemvaleukin in combination with pembrolizumab in platinum-resistant ovarian cancer (ARTISTRY-7) has been initiated.
 - A phase 2 study of nemvaleukin monotherapy in advanced mucosal or cutaneous melanoma (ARTISTRY-6; NCT04830124) is ongoing.

ACKNOWLEDGMENTS AND REFERENCES

Acknowledgments

The authors would like to thank all the patients who are participating in this trial and their families and caregivers. Many thanks to all the investigators and site personnel for their participation in this study. We would also like to acknowledge the inspiring work of Dr Martin "Mac" Cheever, who, until his recent passing, was a leading contributor to the protocol design and execution of the ION-01 study. The trial is sponsored by Alkermes, Inc. Medical writing and editorial support was provided by Parexel and funded by Alkermes, Inc.

References

1. Lopes JE, et al. *J Immunother Cancer.* 2020;8(1):e000673.
2. Boni V, et al. *J Clin Oncol.* 2021;39(Suppl 15):abstr 2513.
3. Hamid O, et al. *J Clin Oncol.* 2021;39(Suppl 15):abstr 2552.
4. Vaishampayan UN, et al. *J Immunother Cancer.* 2019;7(Suppl 1):abstr P447.
5. Powderly JD, et al. *J Immunother Cancer.* 2020;8(Suppl 3):abstr A227.

Copies of this poster obtained through this QR (Quick Response) code are for personal use only and may not be reproduced without permission of Alkermes. For permission, contact: USMedInfo@Alkermes.com

FRED HUTCH
CURES START HERE™

Alkermes®
Patient inspired®

Society for Immunotherapy of Cancer Annual Meeting, November 10-14, 2021, Washington, DC