The Combination of a Mouse Ortholog of ALKS 4230, a Selective Agonist of the Intermediate-Affinity IL-2 Receptor, and the Angiogenesis Inhibitor Lucitanib Enhances Antitumor Activity

Jared E. Lopes,[1] Rachel L. Dusek,[2] Liliane Robillard,[2] Minh Nguyen,[2] Bruce Roth,[1] Bryan Vought,[1] Arthur Liberzon,[1] Raymond J. Winquist,[1] Heather C. Losey[1]

[1]Alkermes, Inc., Waltham, MA; [2]Clovis Oncology, Inc., Boulder, CO

Abstract 2202

INTRODUCTION

- ALKS 4230, a novel cytokine, is an investigational, engineered fusion protein designed to selectively bind to the intermediate-affinity IL-2R. ALKS 4230 is being evaluated as a monotherapy and in combination with pembrolizumab in patients with advanced solid tumors.[1]
- The selectivity of ALKS 4230 is achieved through stable fusion of circularly permuted IL-2 to the IL-2Rα chain, driving antitumor responses via selective activation of CD8+ T cells and NK cells, while avoiding activation of T_{regs}, which express the high-affinity IL-2R.[1]
- High levels of VEGF are associated with an immunosuppressive tumor microenvironment and diminished response to high-dose IL-2.[2]
- Lucitanib is an investigational antiangiogenic multityrosine kinase inhibitor that targets VEGFR1-3, PDGFRα/β, and FGFR1-3.[3]
- We evaluated the antitumor efficacy and mechanism of action of RDB 1462, the mouse ortholog of ALKS 4230, combined with lucitanib in the MC38 mouse syngeneic colon tumor model.

CONCLUSIONS

- The combination of the intermediate-affinity IL-2R-selective cytokine RDB 1462 and the angiogenesis inhibitor lucitanib resulted in durable dose-dependent antitumor efficacy in the MC38 mouse tumor model.
- Tumor infiltrating immune cells and gene expression changes were consistent with increased antitumor immunity, including increased CD8+ T cells and dendritic cells, decreased macrophages, and increased cytolytic gene expression with decreased expression of angiogenesis genes.
- ImmunoSeq analyses demonstrated that T cell receptor diversity increased with RDB 1462 and further increased with the RDB 1462 and lucitanib combination.

METHODS

- C57BL/6 mice were subcutaneously implanted with MC38 cells and, once tumors reached 75 to 125 mm³, were treated with one of the following:
 - RDB 1462 (1.5 mg/kg sc q3d for 3 weeks) + vehicle (po qd) (n = 10)
 - RDB 1462 (3 mg/kg sc q3d for 3 weeks) + vehicle (po qd) (n = 10)
 - Lucitanib (10 mg/kg po qd for 28 days) + vehicle (sc q3d) (n = 10)
 - RDB 1462 (1.5 mg/kg sc q3d for 3 weeks) + lucitanib (10 mg/kg po qd for 28 days) (n = 10)
 - RDB 1462 (3 mg/kg sc q3d for 3 weeks) + lucitanib (10 mg/kg po qd for 28 days) (n = 10)
 - Vehicle only (sc q3d + po qd) (n = 10)
- Individual mice met study endpoint once tumor volumes reached 1000 mm³.
- MC38 tumor cell rechallenge experiment:
 - Mice exhibiting a complete response in the first experiment were rechallenged with a second sc injection of MC38 cells on the opposite flank on day 87.
 - 12 treatment-naïve animals were implanted with MC38 cells to confirm malignancy.
- Additional cohorts of mice were treated with the same regimens (until the day before sampling) for analyses on day 7 (n = 5 each), on day 10 (n = 4 each), and on day 16 (n = 5 each).
- Flow cytometric analyses of samples collected on day 10:
 - Single-cell suspensions were made from tumors and spleens, stained for expression of select markers (Table), and acquired on a BD LSRFortessa flow cytometer.
 - Data were analyzed using FlowJo software (Treestar) version 10.0.7r2.
 - Cell populations were plotted as the percentage of CD45+ cells.
 - Significance was determined using Dunnett's or Tukey's multiple comparison test.

TABLE: Cell Populations and Phenotypic Markers Used for Flow Cytometric Analyses

Cell Population	Phenotypic Markers	Antibody Panel
CD4+ T cells	CD45+CD11b-CD3+CD4+CD8-	CD45, CD3, CD4, CD8, CD11b, CD25, Foxp3, F4/80, CD11c, CD49b, LIVE/DEAD
CD8+ T cells	CD45+CD11b-CD3+CD4-CD8+	
T_{reg} cells	CD45+CD11b-CD3+CD4+CD25+Foxp3+	
Macrophages	CD45+CD3-CD11b+F4/80+	
Dendritic cells	CD45+CD3-CD11b-CD11c+F4/80-	

- RNA-Seq analyses of samples collected on day 10:
 - RNA was isolated from tumors stored in RNAlater using a combination of a Trizol extraction and a Qiagen RNA Mini kit. RNA samples were quantified on a Qubit 4 Fluorometer using the RNA HS assay kit.
 - Gene expression was measured on a ThermoFisher GeneStudio S5 using the Ampliseq mouse Transcriptome kit.
- Gene set enrichment analysis:
 - GSEA[5] assessed enrichment of 167 KEGG pathways (v7.0 MSigDB) in each condition. The heatmap shows 40 most significant GSEA results.
- T cell receptor repertoire analyses of samples collected on days 7 and 16:
 - Analyses were performed on DNA isolated from tumors using the ImmunoSeq platform by Adaptive Biotechnologies.

RESULTS

FIGURE 1: Combination of RDB 1462 and Lucitanib Results in Complete Responses and Enhanced Survival Compared With Monotherapy Treatments

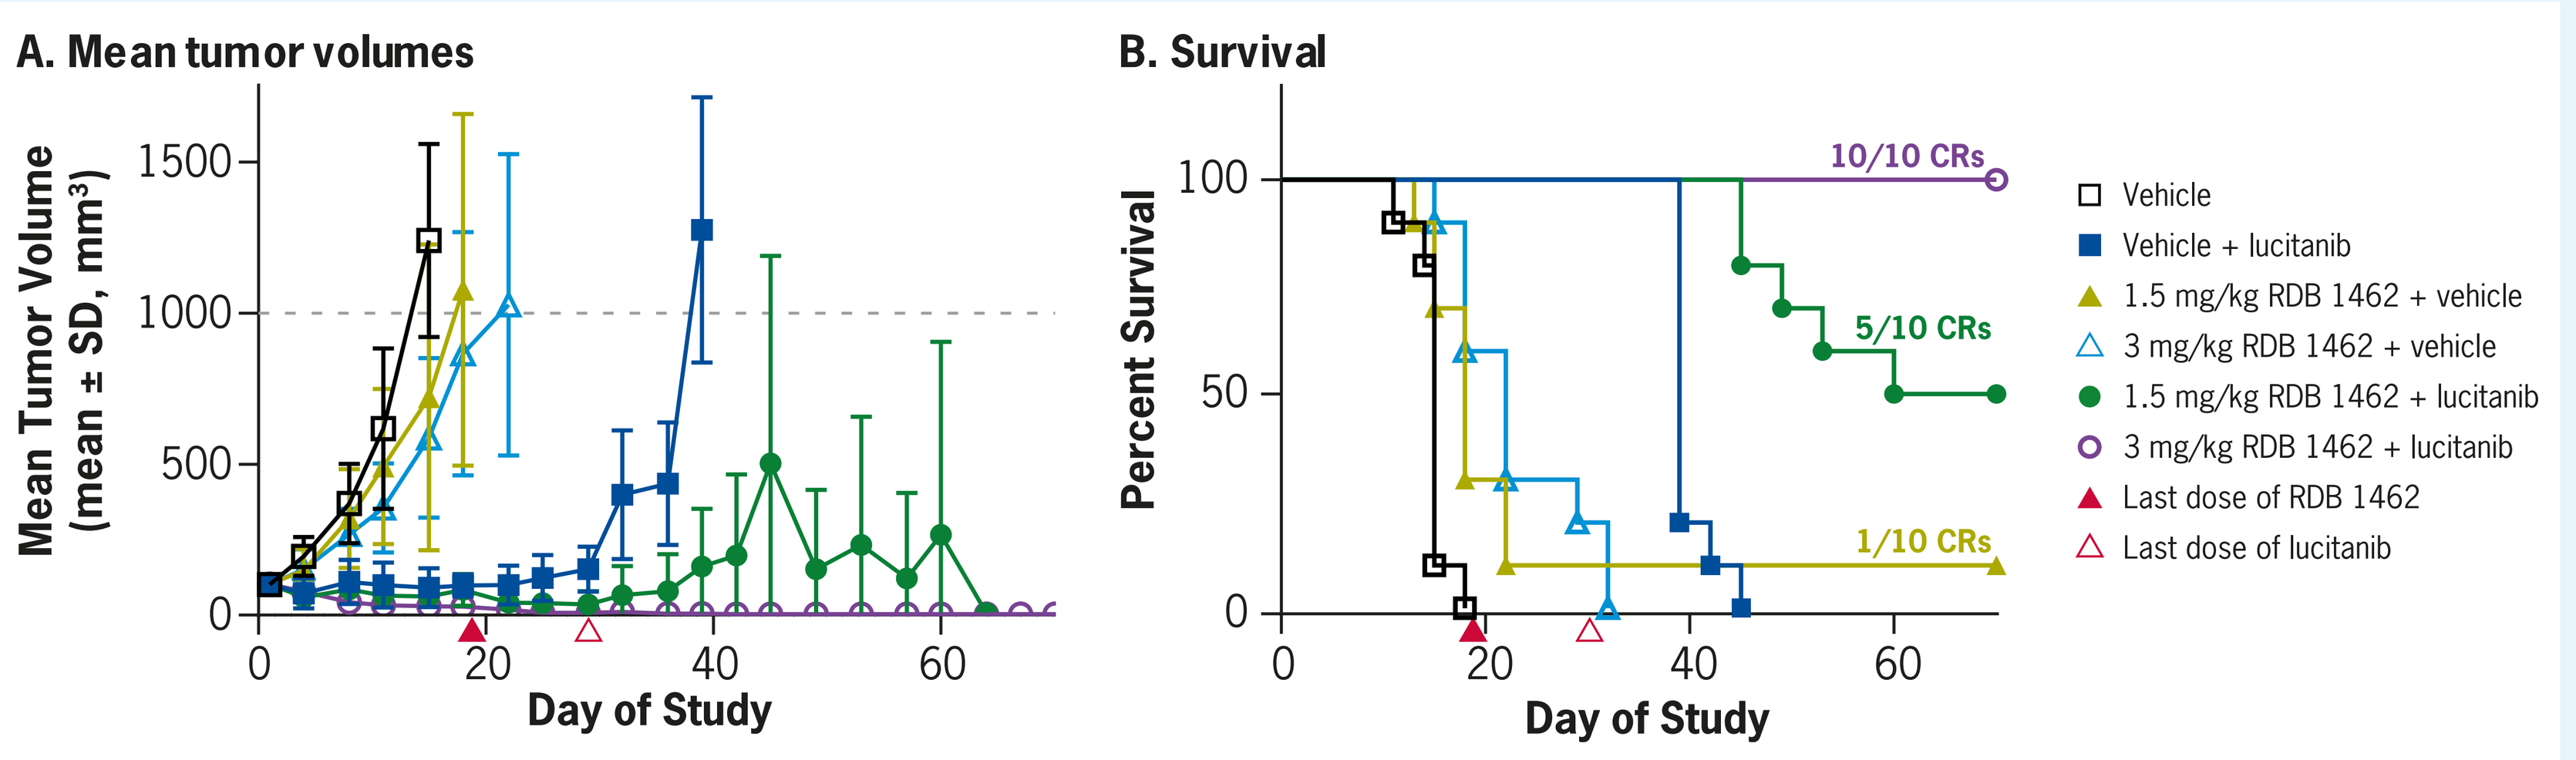
A. Mean tumor volumes
Mean Tumor Volume (mean ± SD, mm³)
Day of Study
B. Survival
Percent Survival
Day of Study
10/10 CRs
5/10 CRs
1/10 CRs
Vehicle
Vehicle + lucitanib
1.5 mg/kg RDB 1462 + vehicle
3 mg/kg RDB 1462 + vehicle
1.5 mg/kg RDB 1462 + lucitanib
3 mg/kg RDB 1462 + lucitanib
Last dose of RDB 1462
Last dose of lucitanib

- Both RDB 1462 dose combinations and lucitanib monotherapy resulted in 100% TGI, with TGI rates of 25% and 51% observed following RDB 1462 monotherapy (1.5 and 3 mg/kg, respectively).
- Both dose combinations of RDB 1462 and lucitanib extended survival (MST 68.5 days and > 77 days; low- and high-dose combinations, respectively) versus RDB 1462 (MST 18 and 22 days, respectively) and lucitanib (39 days) monotherapy.
 - All survival curves were significantly different from that for vehicle group (Mantel-Cox, $P < 0.05$).
- Rechallenge of all complete responders (CRs) demonstrated protection mediated by immunological memory (data not shown).

FIGURE 2: RDB 1462, as Monotherapy and in Combination With Lucitanib, Selectively Expands CD8+ T Cells in Tumors

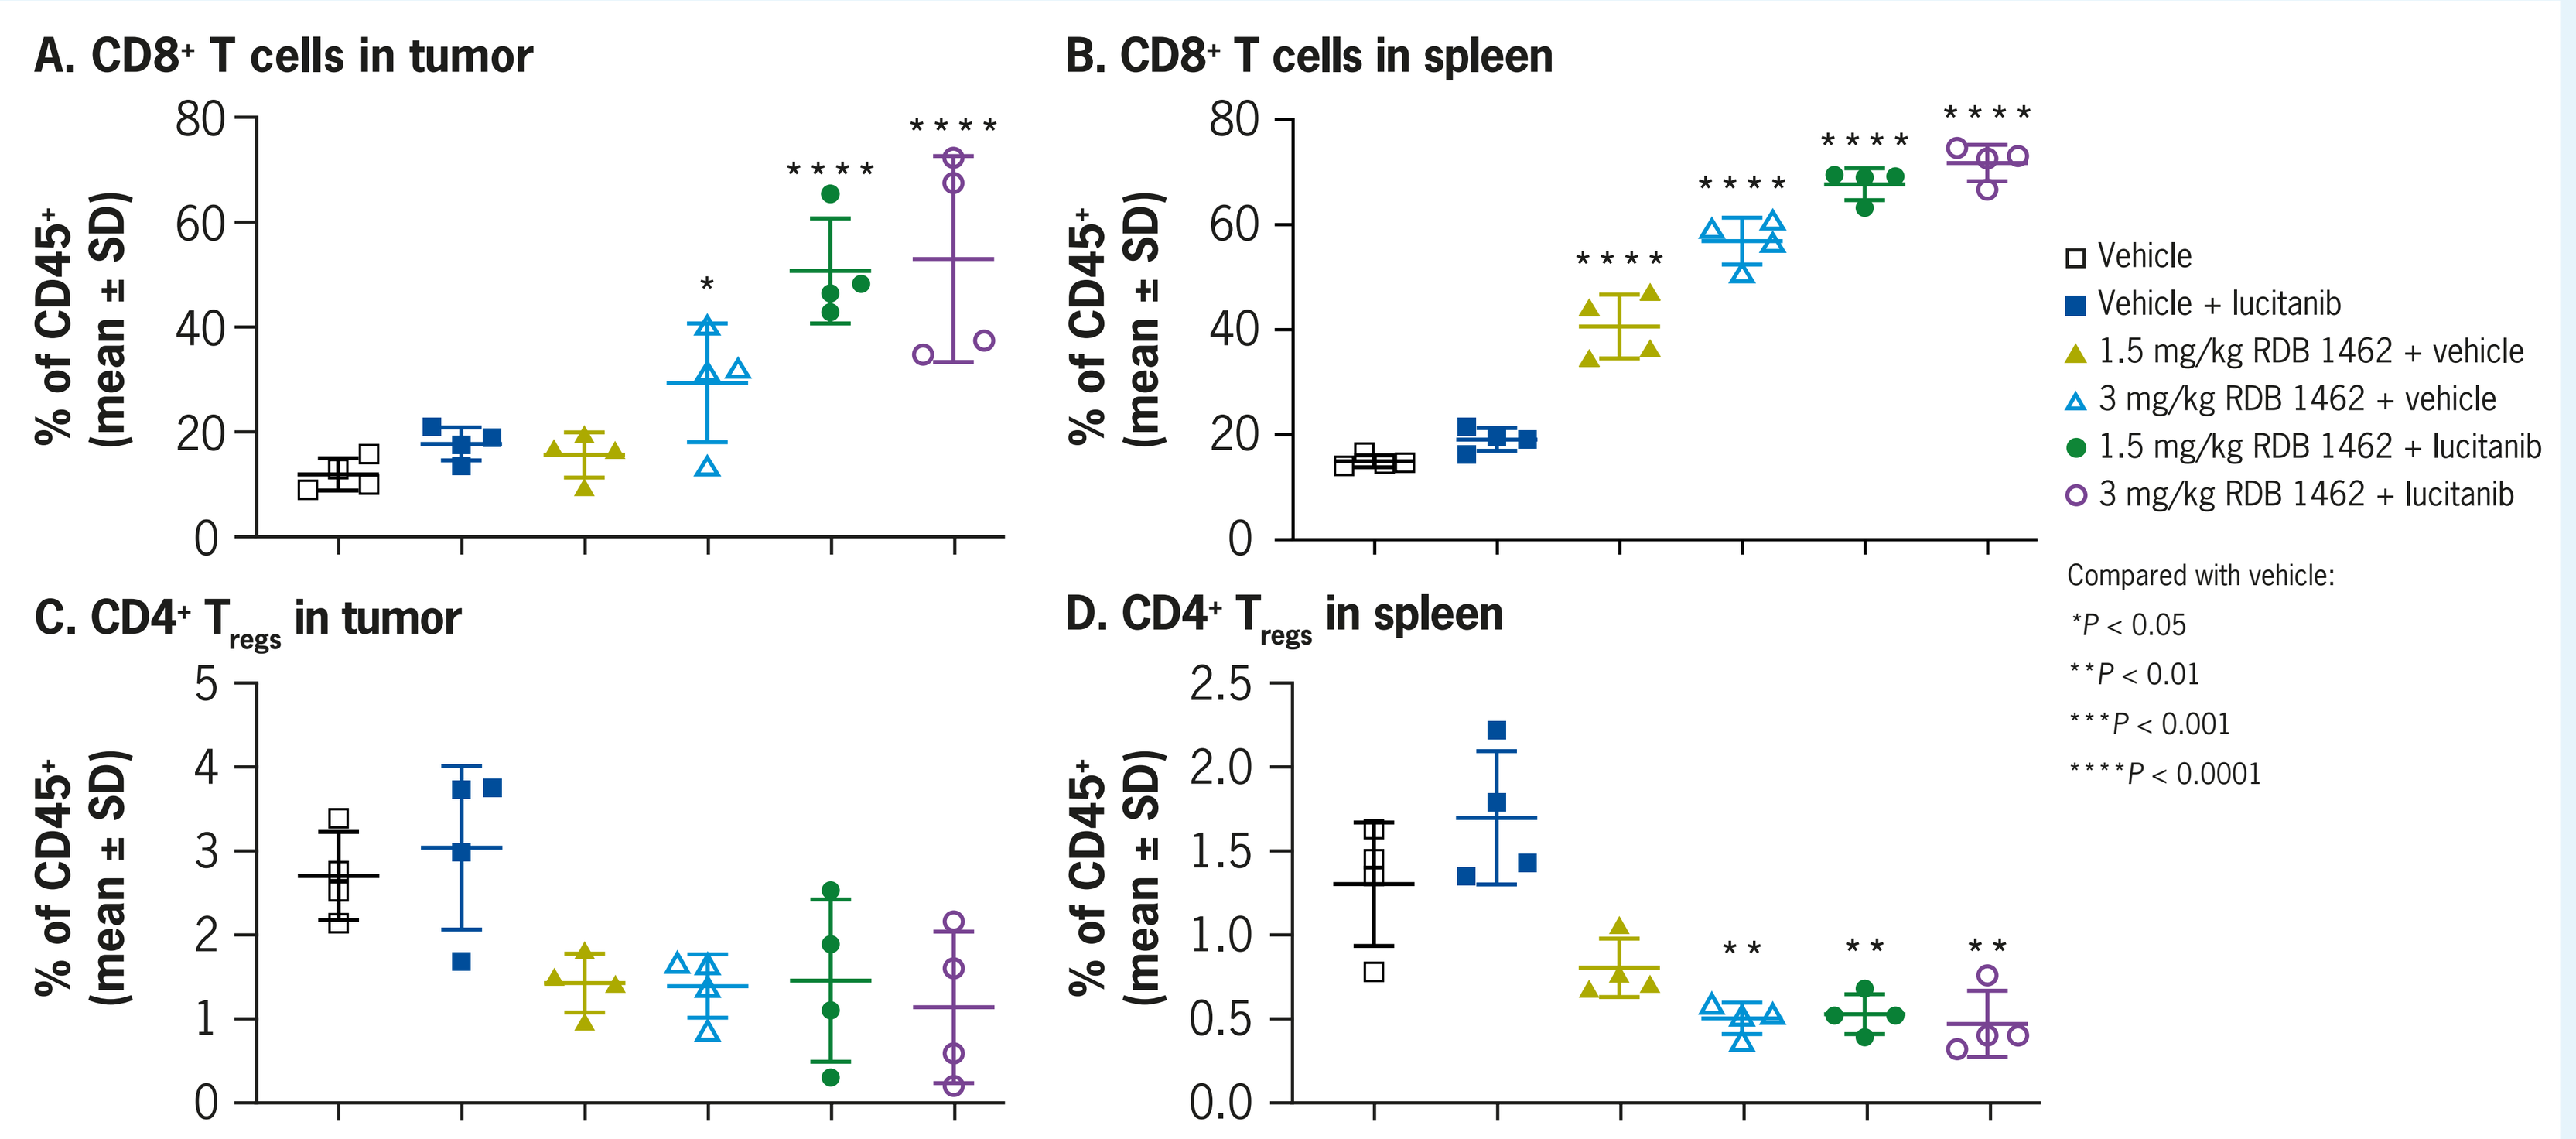
A. CD8+ T cells in tumor
B. CD8+ T cells in spleen
C. CD4+ Tregs in tumor
D. CD4+ Tregs in spleen
% of CD45+ (mean ± SD)
Vehicle
Vehicle + lucitanib
1.5 mg/kg RDB 1462 + vehicle
3 mg/kg RDB 1462 + vehicle
1.5 mg/kg RDB 1462 + lucitanib
3 mg/kg RDB 1462 + lucitanib
Compared with vehicle:
*P < 0.05
**P < 0.01
***P < 0.001
****P < 0.0001

- RDB 1462 monotherapy induced significant CD8+ T cell expansion in tumor and spleen versus vehicle ($P < 0.05$), with no increase in CD4+ T_{regs}.
- The effect of RDB 1462 on CD8+ T cells was enhanced when it was given in combination with lucitanib.

FIGURE 3: Combination of RDB 1462 and Lucitanib Resulted in Increased Dendritic Cells but Fewer Tumor-Associated Macrophages Compared With Vehicle

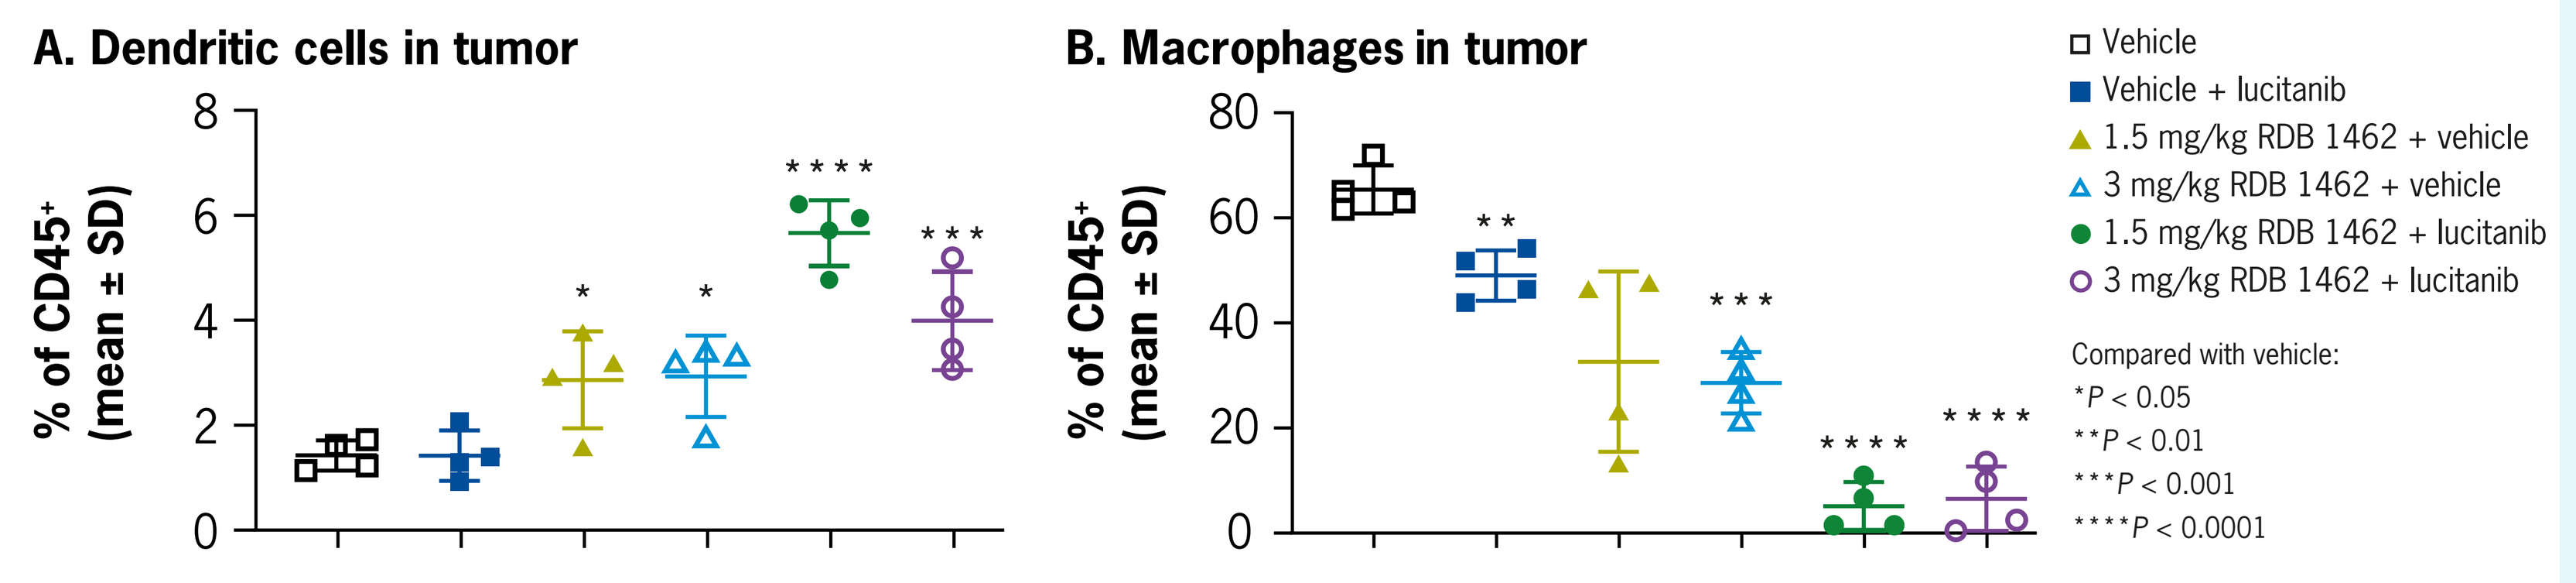
A. Dendritic cells in tumor
B. Macrophages in tumor
% of CD45+ (mean ± SD)
Vehicle
Vehicle + lucitanib
1.5 mg/kg RDB 1462 + vehicle
3 mg/kg RDB 1462 + vehicle
1.5 mg/kg RDB 1462 + lucitanib
3 mg/kg RDB 1462 + lucitanib
Compared with vehicle:
*P < 0.05
**P < 0.01
***P < 0.001
****P < 0.0001

- The combination of RDB 1462 and lucitanib resulted in a significant decrease in macrophages and a significant increase in intratumoral dendritic cells compared to vehicle.

Abbreviations: ANOVA, analysis of variance; *Esm1*, endothelial cell–specific molecule 1; FDR, false discovery rate; FGFR, fibroblast growth factor receptor; GSEA, gene set enrichment analysis; IL-2R, interleukin-2 receptor; KEGG, Kyoto Encyclopedia of Genes and Genomes; MST, mean survival time; PDGFR, platelet-derived growth factor receptor; po, orally; q3d, every 3 days; qd, every day; RPM, reads per million; sc, subcutaneously; SEM, standard error of the mean; TGI, tumor growth inhibition; T_{regs}, regulatory T cells; VEGF, vascular endothelial growth factor; VEGFR, VEGF receptor.

References: **1**. Losey HC, et al. Presented at the AACR Annual Meeting; April 1-5, 2017; Washington, DC. Abs 591. **2**. Ott PA, et al. *Front Oncol.* 2015;5:202. **3**. Dusek RL, et al. *Cancer Res.* 2019;79(13 suppl):1214. **4**. Rooney MS, et al. *Cell.* 2015;160:48-61. **5**. Subramanian A, et al. *PNAS* 2005;102(43):15545-15550.

FIGURE 4: Combination of RDB 1462 and Lucitanib Results in Changes in Gene Expression in Tumors Not Observed With Either Monotherapy, as Determined by RNA-Seq Analyses

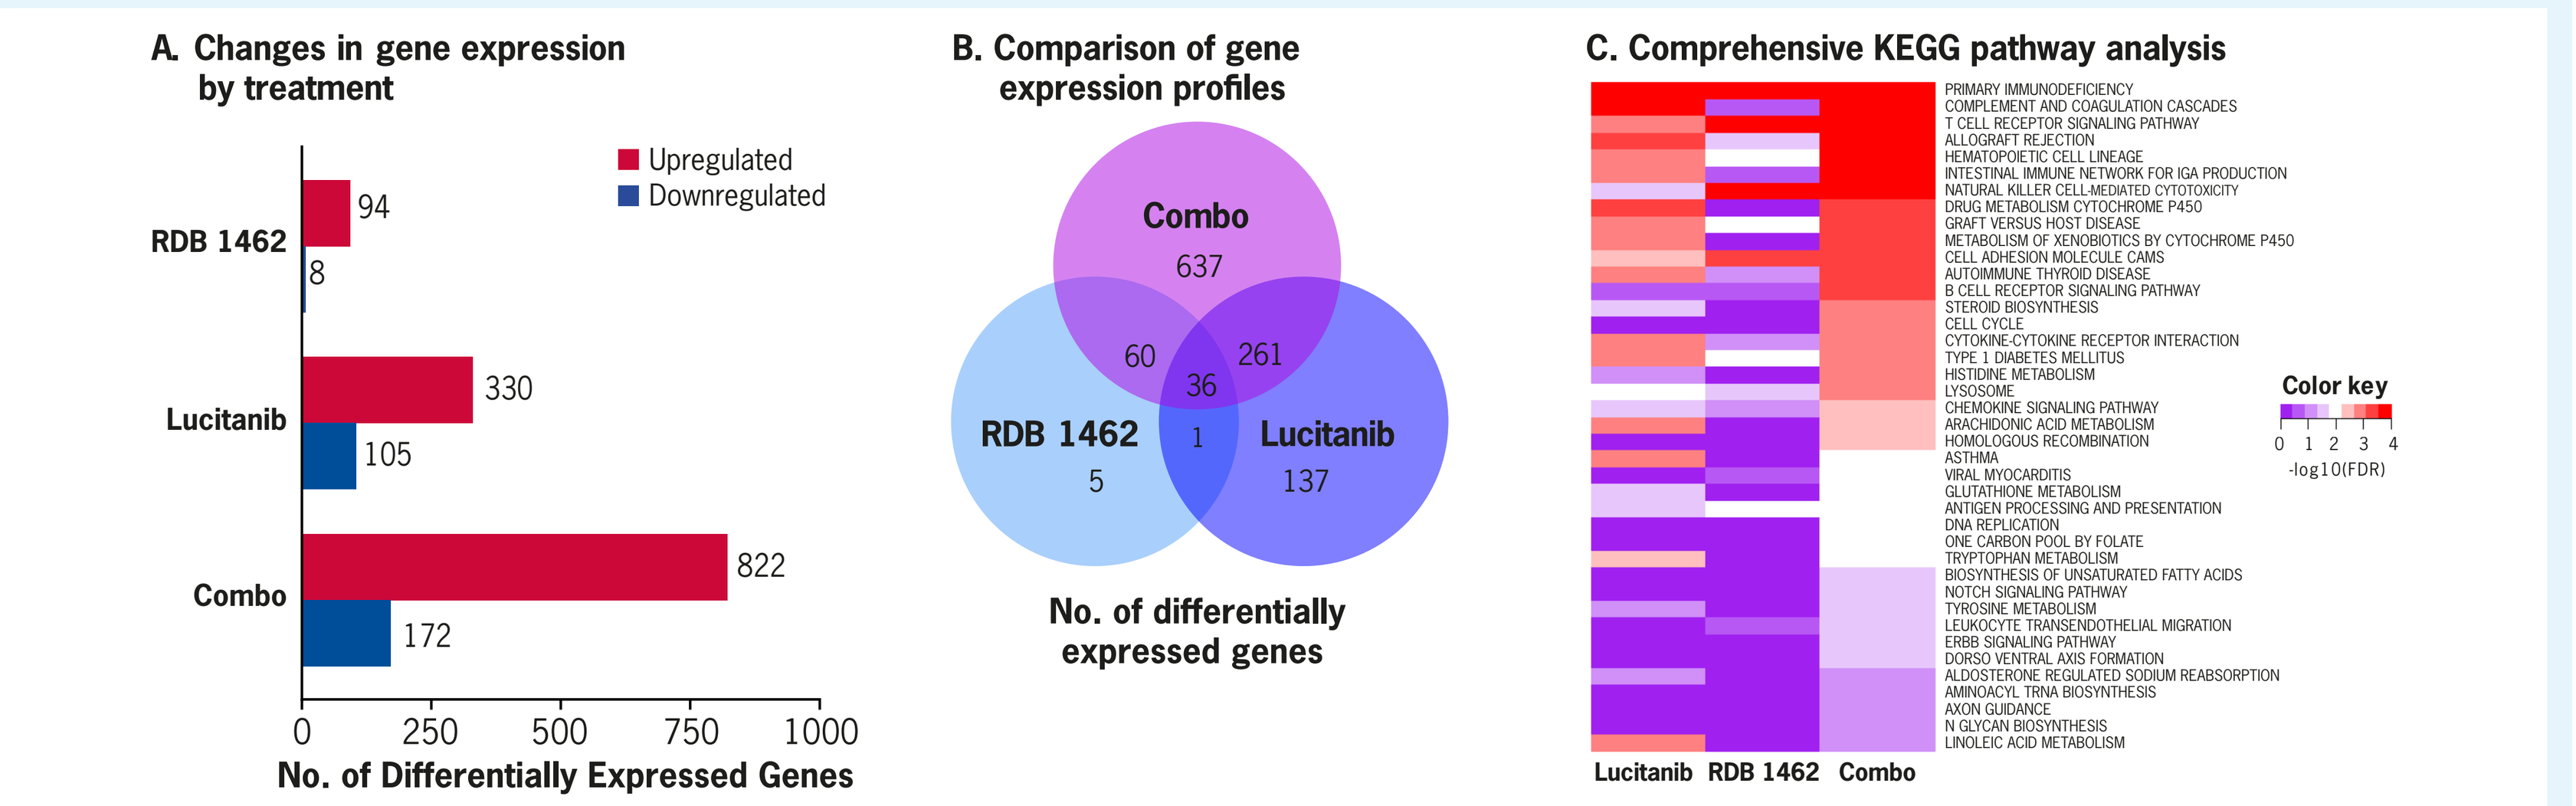
A. Changes in gene expression by treatment
Upregulated
Downregulated
RDB 1462
94
8
Lucitanib
330
105
Combo
822
172
No. of Differentially Expressed Genes
B. Comparison of gene expression profiles
Combo
637
60
261
36
1
RDB 1462
5
Lucitanib
137
No. of differentially expressed genes
C. Comprehensive KEGG pathway analysis
PRIMARY IMMUNODEFICIENCY
COMPLEMENT AND COAGULATION CASCADES
T CELL RECEPTOR SIGNALING PATHWAY
ALLOGRAFT REJECTION
HEMATOPOIETIC CELL LINEAGE
INTESTINAL IMMUNE NETWORK FOR IGA PRODUCTION
NATURAL KILLER CELL MEDIATED CYTOTOXICITY
DRUG METABOLISM CYTOCHROME P450
GRAFT VERSUS HOST DISEASE
METABOLISM OF XENOBIOTICS BY CYTOCHROME P450
CELL ADHESION MOLECULE CAMS
AUTOIMMUNE THYROID DISEASE
B CELL RECEPTOR SIGNALING PATHWAY
STEROID BIOSYNTHESIS
CELL CYCLE
CYTOKINE CYTOKINE RECEPTOR INTERACTION
TYPE 1 DIABETES MELLITUS
HISTIDINE METABOLISM
LYSOSOME
CHEMOKINE SIGNALING PATHWAY
ARACHIDONIC ACID METABOLISM
HOMOLOGOUS RECOMBINATION
ASTHMA
VIRAL MYOCARDITIS
GLUTATHIONE METABOLISM
ANTIGEN PROCESSING AND PRESENTATION
DNA REPLICATION
ONE CARBON POOL BY FOLATE
TRYPTOPHAN METABOLISM
BIOSYNTHESIS OF UNSATURATED FATTY ACIDS
NOTCH SIGNALING PATHWAY
TYROSINE METABOLISM
LEUKOCYTE TRANSENDOTHELIAL MIGRATION
ERBB SIGNALING PATHWAY
DORSO VENTRAL AXIS FORMATION
ALDOSTERONE REGULATED SODIUM REABSORPTION
AMINOACYL TRNA BIOSYNTHESIS
AXON GUIDANCE
N GLYCAN BIOSYNTHESIS
LINOLEIC ACID METABOLISM
Lucitanib RDB 1462 Combo
Color key
-log10(FDR)

- Genes showing a 2-fold change or greater relative to vehicle ($P < 0.05$; ANOVA) are represented in panels A and B.
- The combination of 3 mg/kg RDB 1462 and lucitanib resulted in an increase in the number of differentially expressed genes (panel A) with a diverse profile (panel B) versus 3 mg/kg RDB 1462 monotherapy or lucitanib monotherapy.
- The heatmap in panel C shows GSEA of KEGG pathways. The 40 most enriched pathways are shown. Color key indicates the negative log10 of P values (FDR). Combination treatment resulted in more significant enrichment of pathways associated with monotherapy treatment as well as significant enrichment of unique pathways.

FIGURE 5: Both RDB 1462 and Lucitanib Monotherapy Treatments Induce Cytolytic Gene Expression and T Cell Activation, Which Is Complemented by Lucitanib-Mediated Regulation of VEGF Pathway Genes

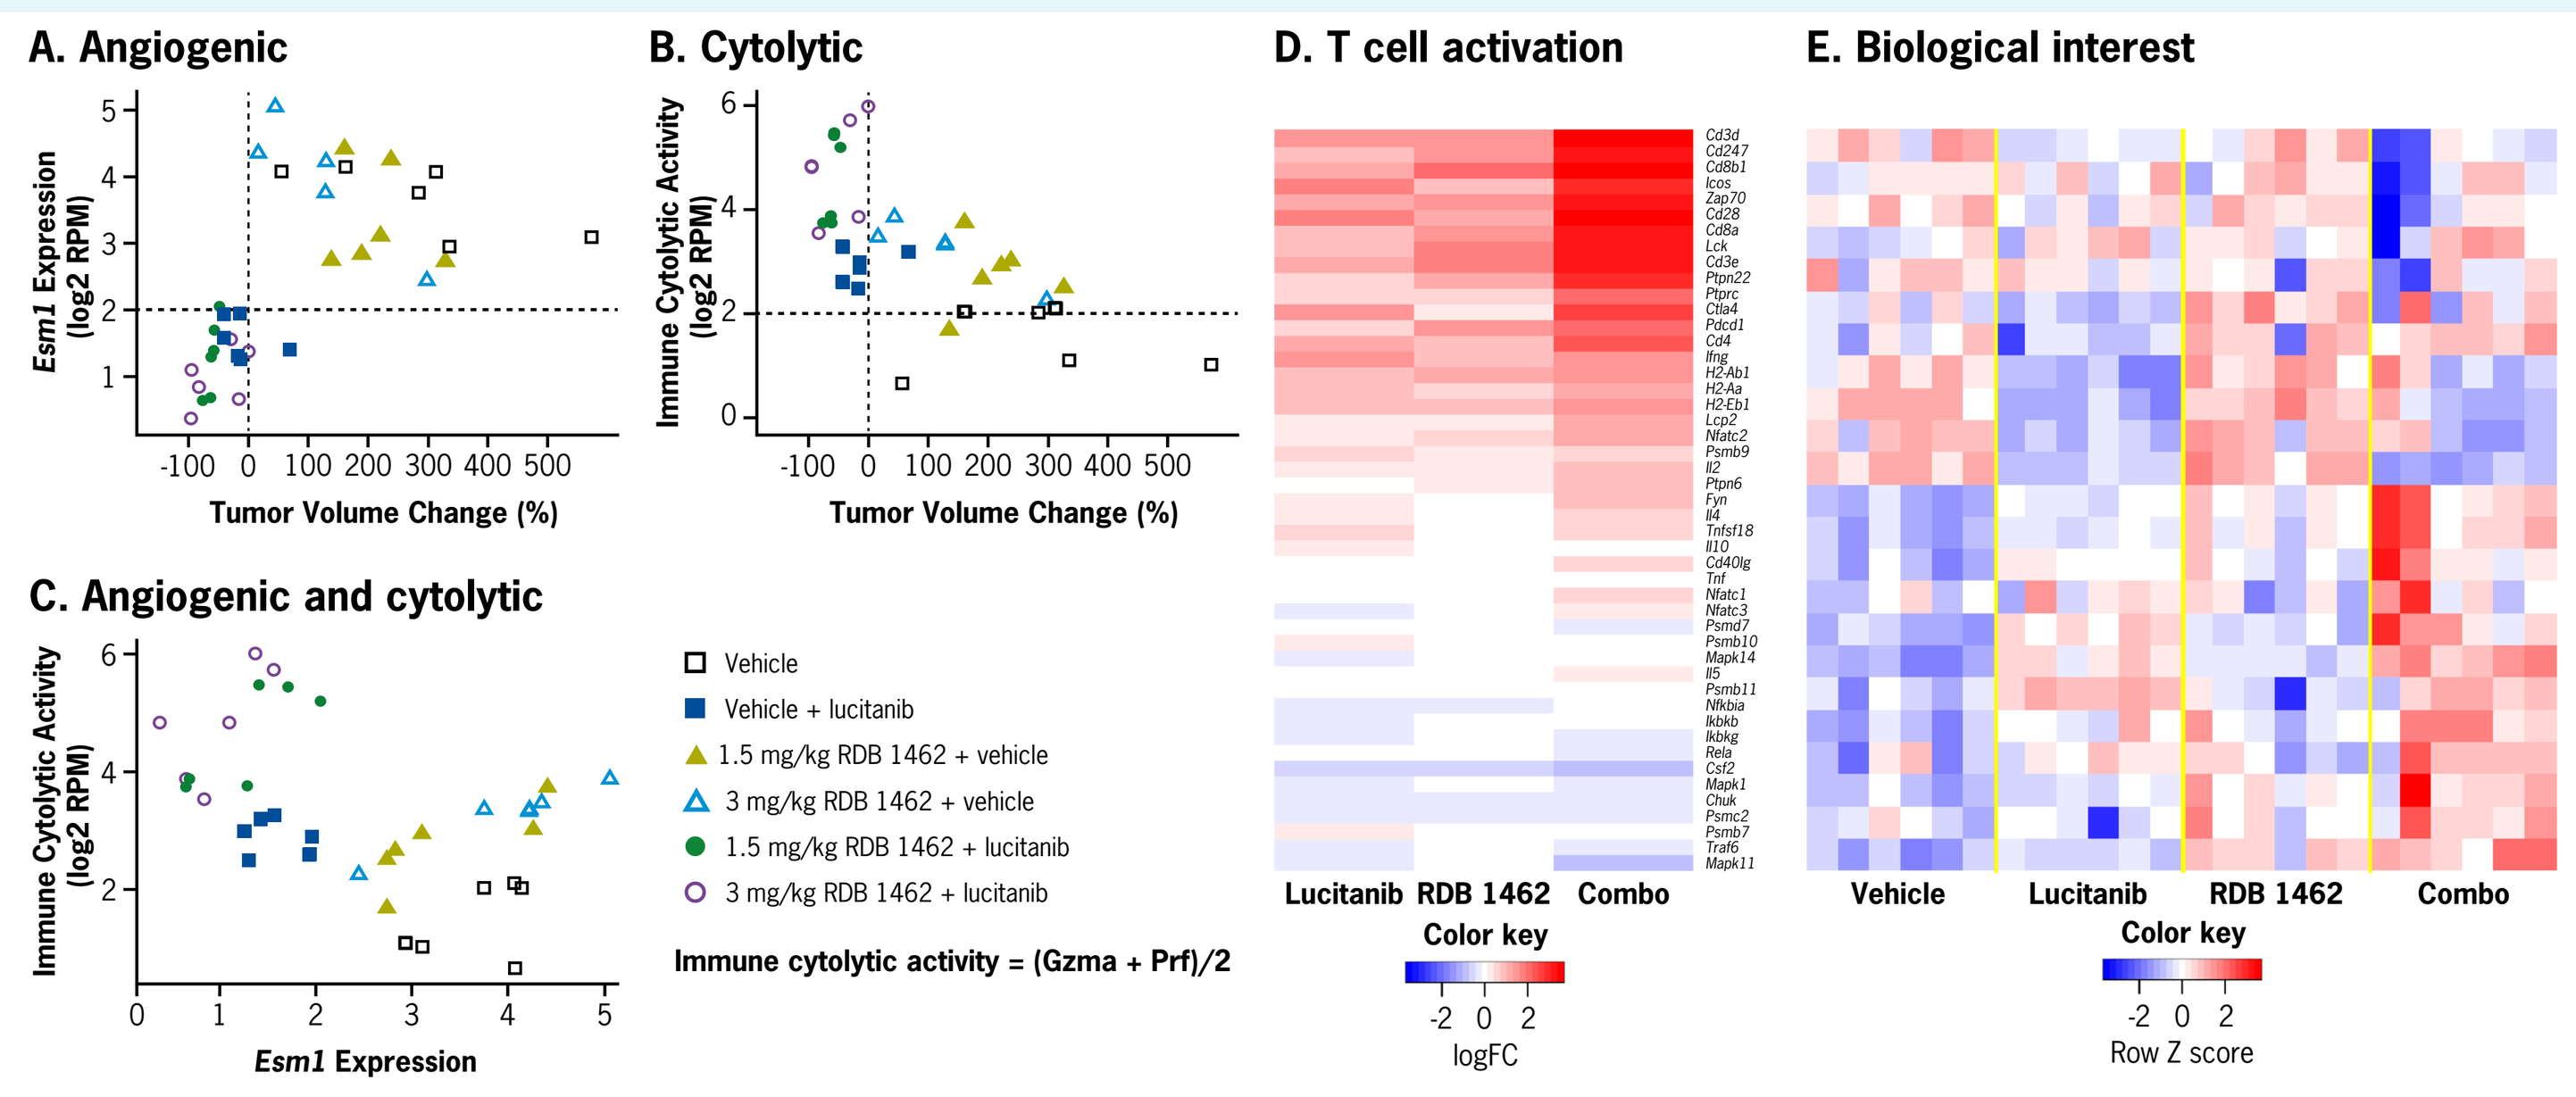
A. Angiogenic
Esm1 Expression (log2 RPM)
Tumor Volume Change (%)
B. Cytolytic
Immune Cytolytic Activity (log2 RPM)
Tumor Volume Change (%)
C. Angiogenic and cytolytic
Immune Cytolytic Activity (log2 RPM)
Esm1 Expression
Vehicle
Vehicle + lucitanib
1.5 mg/kg RDB 1462 + vehicle
3 mg/kg RDB 1462 + vehicle
1.5 mg/kg RDB 1462 + lucitanib
3 mg/kg RDB 1462 + lucitanib
Immune cytolytic activity = (Gzma + Prf)/2
D. T cell activation
Lucitanib RDB 1462 Combo
Color key
logFC
E. Biological interest
Vehicle Lucitanib RDB 1462 Combo
Color key
Row Z score

FDR q-values			
	Treatment		
Gene	Lucitanib	RDB 1462	Combo
Cd38	0.734	1	0.2999
Cd93	0.6479	1	0.8845
Cd8b1	0.4668	1	4.00E-04
Flt1	0.3231	1	0.0086
Cd163	0.8298	1	0.233
Kdr	0.0386	1	0.3371
Gzma	0.6189	0.6197	2.00E-04
Flt4	0.0035	1	0.0106
Esm1	0.002	1	2.12E-05
Cd4	0.3192	1	0.0031
Cd8a	0.5289	1	4.00E-04
Pdcd1	0.7532	1	0.0086
Msr1	0.8369	1	0.9301
Mrc1	0.5217	1	0.0344
Ly6a	0.0571	1	0.0352
Mertk	0.9202	1	0.1365
Angpt2	0.1775	1	0.1541
Vegfb	0.9991	1	0.2794
Prf1	0.0013	0.6743	2.48E-07
Havcr2	0.2614	1	0.0013
Vegfa	0.7274	1	0.0344
Foxp3	0.0477	1	2.00E-04
Aspm	0.9803	1	0.2044

- Reduction in expression of *Esm1* (a marker of VEGF activity) correlated with decreased tumor volume in lucitanib mono- and combination therapy–treated animals (panel A).
- RDB 1462 and lucitanib monotherapy induced cytolytic gene expression while combination treatment resulted in greater cytolytic gene expression, which appears to be inversely correlated with changes in tumor volume (panel B).
- Combination treatment resulted in greater cytolytic gene expression as well as reduction in *Esm1* expression (panel C).
- Upregulation of selected genes involved in T cell activation was observed (panel D); color key indicates log2 fold change relative to vehicle.
- Genes with activity in pathways of interest are regulated by combination treatment as indicated by significant q values (FDR; panel E).

FIGURE 6: Combination of RDB 1462 and Lucitanib Results in Increased T Cell Receptor Diversity, Increased T Cell Fraction, and Distinct T Cell Receptor Repertoires in the Tumors of Treated Mice

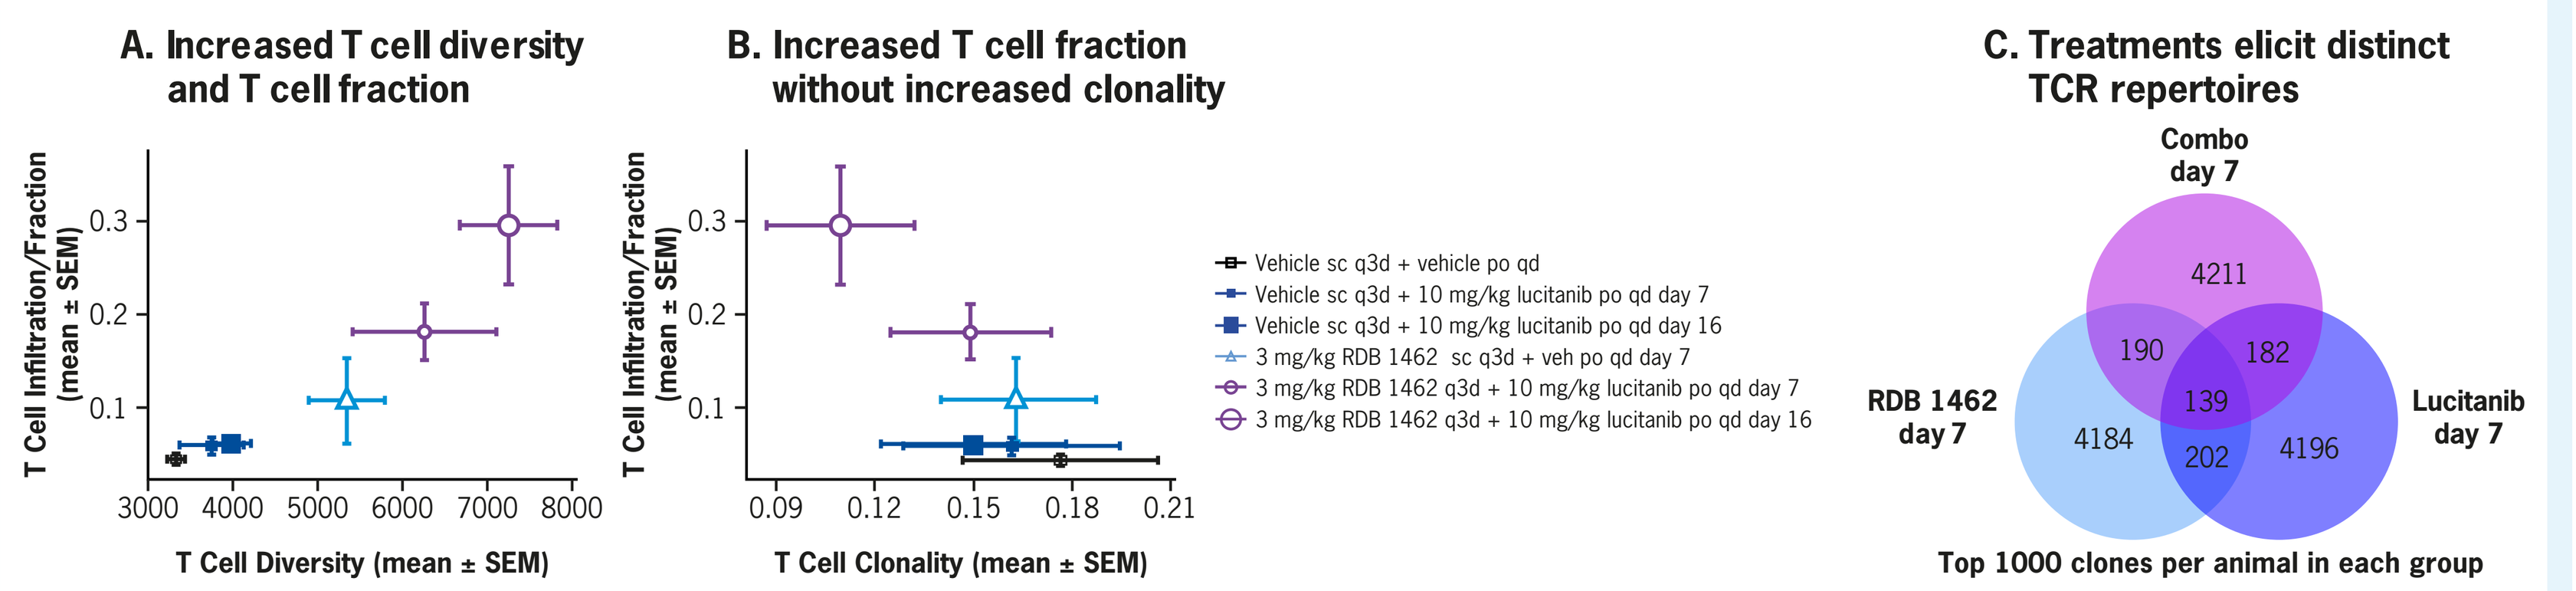
A. Increased T cell diversity and T cell fraction
T Cell Infiltration/Fraction (mean ± SEM)
T Cell Diversity (mean ± SEM)
B. Increased T cell fraction without increased clonality
T Cell Clonality (mean ± SEM)
Vehicle sc q3d + vehicle po qd
Vehicle sc q3d + 10 mg/kg lucitanib po qd day 7
Vehicle sc q3d + 10 mg/kg lucitanib po qd day 16
3 mg/kg RDB 1462 sc q3d + veh po qd day 7
3 mg/kg RDB 1462 q3d + 10 mg/kg lucitanib po qd day 7
3 mg/kg RDB 1462 q3d + 10 mg/kg lucitanib po qd day 16
C. Treatments elicit distinct TCR repertoires
Combo day 7
4211
190
182
139
4184
202
4196
RDB 1462 day 7
Lucitanib day 7
Top 1000 clones per animal in each group

- T cell fraction, T cell receptor diversity, and Simpson's T cell clonality were determined for tumors collected from mice with the indicated treatments on days 7 and 16. Vehicle-treated and 3 mg/kg RDB 1462 monotherapy groups were excluded from analysis on day 16 due to a number of animals reaching endpoint (Panels A and B).
 - Together, these data suggest the combination of RDB 1462 and lucitanib results in the broadening of the anti-tumor T cell response.
- Top 1000 T cell receptor sequences from each tumor in the indicated groups were compared to evaluate the potential overlap in T cell receptor repertoires (Panel C).

Copies of this poster obtained through this QR (Quick Response) code are for personal use only and may not be reproduced without permission of Alkermes. For permission, contact: USMedInfo@Alkermes.com

AACR Virtual Annual Meeting II, June 22-24, 2020